P0678

High-efficiency homology-directed insertion into the genome using ARCUS nucleases.

Adam Mischler, Laura Christian-Resly, Alex Vogel, Alan Tubbs, JoAnn Hux, Jason Harris, Ginger Tomberlin, Ian MacDonald, Jeff Sunman, Janel Lape, J. Jeff Smith, Aaron J. Martin

All authors are employees of Precision BioSciences, Inc.

INTRODUCTION

There are now many gene editing tools poised to enter the clinic, however a majority of gene editing modalities are focused on deleting or knocking-out genes, thereby limiting broad therapeutic applicability. For instance, base editors are accomplished at switching out a particular base but cannot make all forms of base changes, nor do they support large gene deletions or perform more sophisticated tasks like gene insertions which offer broader therapeutic potential for diseases that require a gain in function. **Stimulating gene editing by homology directed repair (HDR) has the potential advantage of being able to accomplish any type of edit, as defined by the repair template, but gene editing enzymes that support efficient HDR are a relative rarity.**

Homing endonucleases are a family of proteins that have evolved to safely insert a copy of their gene into a genome using HDR. ARCUS nucleases are engineered from the homing endonuclease I-CreI and have programmable sequence specificity. The following work demonstrates that ARCUS nucleases maintain the ability of a homing endonuclease to drive efficient DNA repair by HDR. In T lymphocytes, we observed that HDR-mediated insertion of a DNA template could be achieved in greater than 85% of cells when the template was delivered by AAV6. Following cleavage of its DNA target site, ARCUS nucleases leave behind a double-stranded break (DSB) featuring a 4-base pair, 3′ overhang. We demonstrated that removing this overhang (with exogenous exonuclease activity, or by introducing a DSB with a blunt-cutting endonuclease) resulted in greatly reduced insertion efficiency. We further explored the importance of the cut generated by ARCUS, as well as the impact of homology arms and the contributions of the cellular DNA repair machinery, as they pertain to genomic insertion.

Using HDR, we demonstrated the ability of ARCUS to support the full range of currently understood DNA editing approaches including all combinations of base changes, introducing small, specific gene deletions, small and large gene insertions, and even the ability to repair and replace large segments of DNA within the genome. Collectively, we show that the characteristic enzymology of ARCUS nucleases drives high rates of HDR and that this can be leveraged to execute a multitude of highly precise gene editing functions for therapeutic benefit.

APPROACH

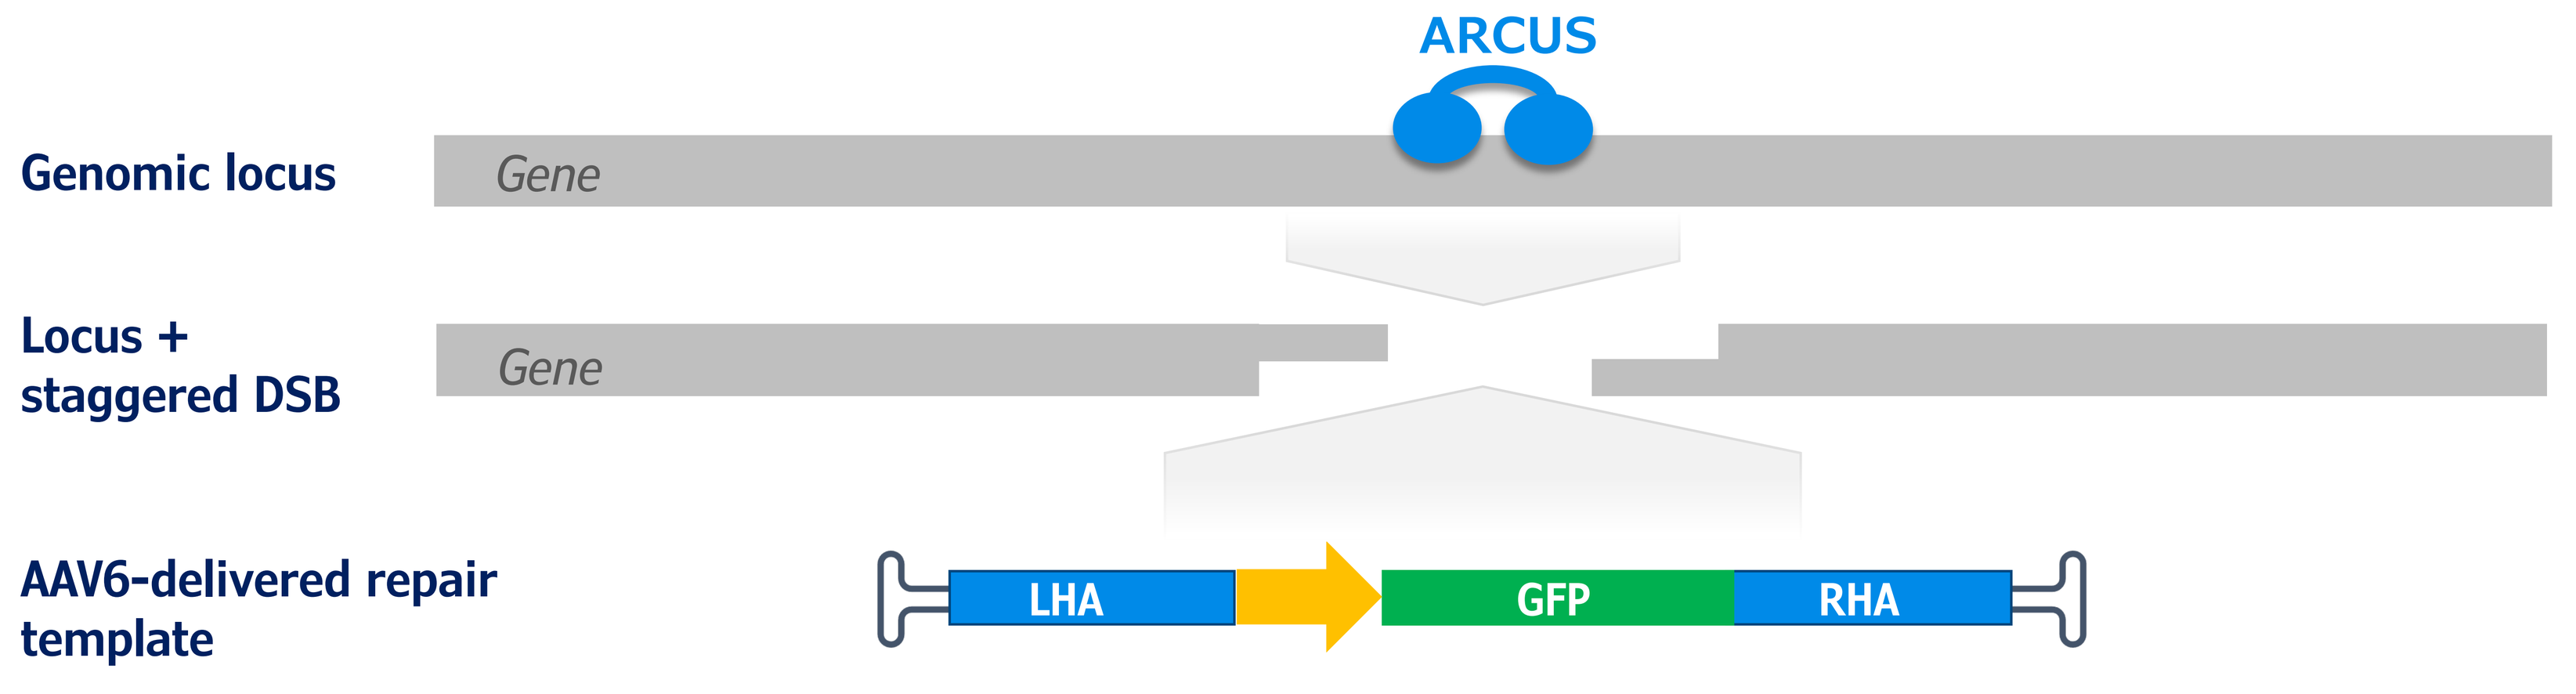
ARCUS
Genomic locus
Gene
Locus +
staggered DSB
Gene
AAV6-delivered repair
template
LHA
GFP
RHA

- We demonstrate that we can accomplish highly efficient gene insertion in T-cells and PHHs (Figures 1 and 2 – below).
- We demonstrate that we accomplish this efficient insertion by HDR using variations in repair template, cut DNA ends, and inhibitors of various DNA repair mechanisms (Figures 3 and 4)
- Using HDR, we demonstrate base change capacity (all 12 possible alternatives), small and large insertions, small deletions, and large replacements in the genome (Figures 5 and 6).

RESULTS

ARCUS enables efficient targeted integration

Figure 1: ARCUS DSBs and AAV6 repair templates support efficient transgene integration.

Human T lymphocytes were edited with an ARCUS nuclease specific for the T cell receptor, alpha chain constant region (TRAC) locus. Cells were given vector alone (left), ARCUS mRNA alone (center) or both (right). Transgene expression requires integration and is observed in approximately 90% of edited T cells. Cells received 500ng of mRNA and 2.0 x 10^4 viral genomes (v.g.) per cell.

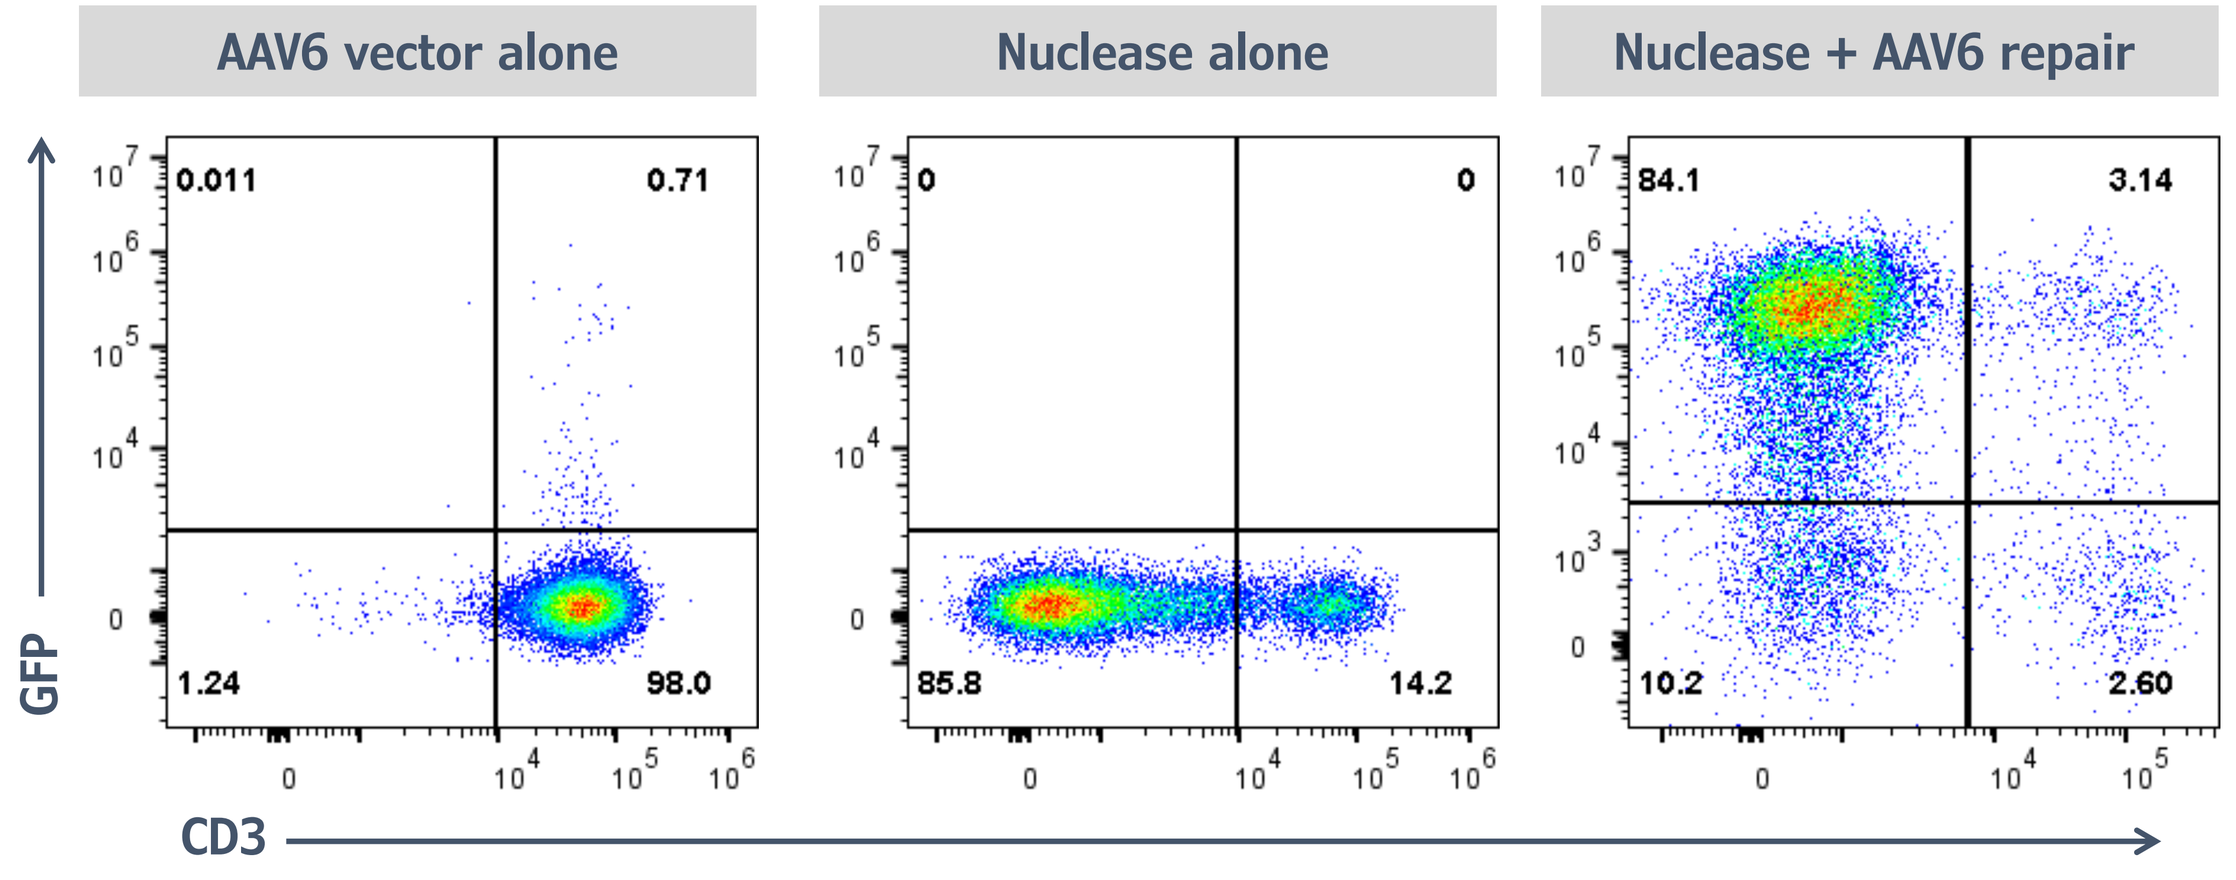
AAV6 vector alone
Nuclease alone
Nuclease + AAV6 repair
0.011
0.71
1.24
98.0
0
0
85.8
14.2
84.1
3.14
10.2
2.60
GFP
CD3

Figure 2: ARCUS enables efficient transgene integration into noncycling primary human hepatocytes.

Human hepatocytes (PHH) were edited with an ARCUS nuclease targeting hydroxyacid oxidase (HAO1), delivered via LNP and were transduced with an AAV carrying a repair template. Seven days later, ddPCR was performed to quantify alleles with an indel (50-90%) or with a transgene insert (39%).

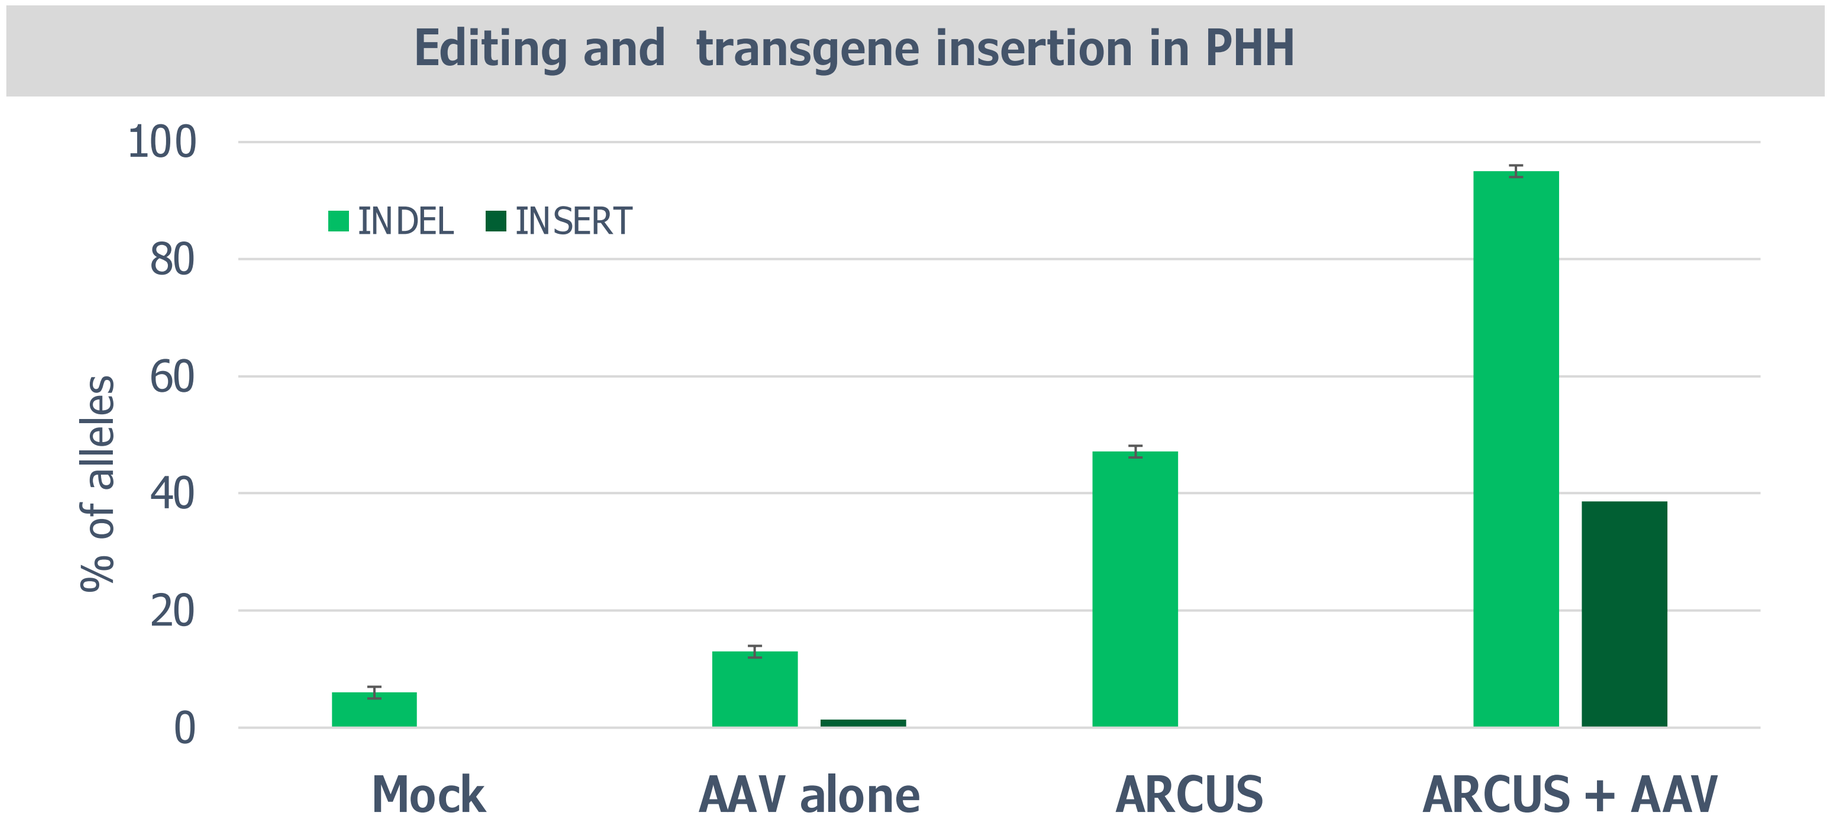
Editing and transgene insertion in PHH
INDEL
INSERT
% of alleles
Mock
AAV alone
ARCUS
ARCUS + AAV

ARCUS DSBs are repaired via HDR

Figure 3: ARCUS DSBs are repaired by HDR.

A) T lymphocytes were edited at the TRAC locus and transduced with AAV6 vectors containing homology arms on both sides of the transgene, or on only left side (LHA), or only right sides (RHA). Other vectors displayed no homology arms, or microhomology arms. **B)** GFP integration frequency and signal intensity were highest when two homology arms were present. Non-homologous inserts exhibit impaired GFP expression strength. **C)** Nanopore long-read sequencing determined that vector ends displaying a homology arm repaired the DSB via HDR while ends lacking homology repaired via end-joining.

A)

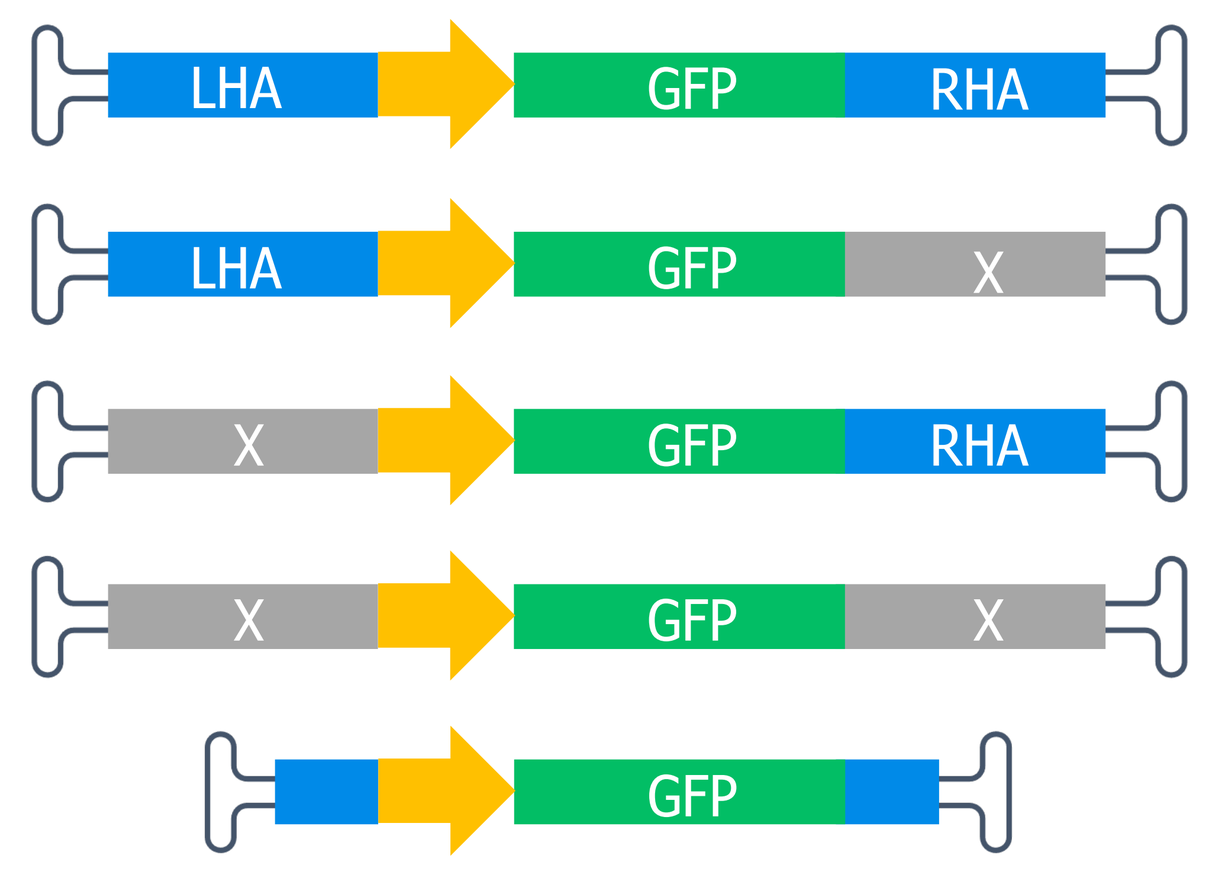
LHA
GFP
RHA
X

B)

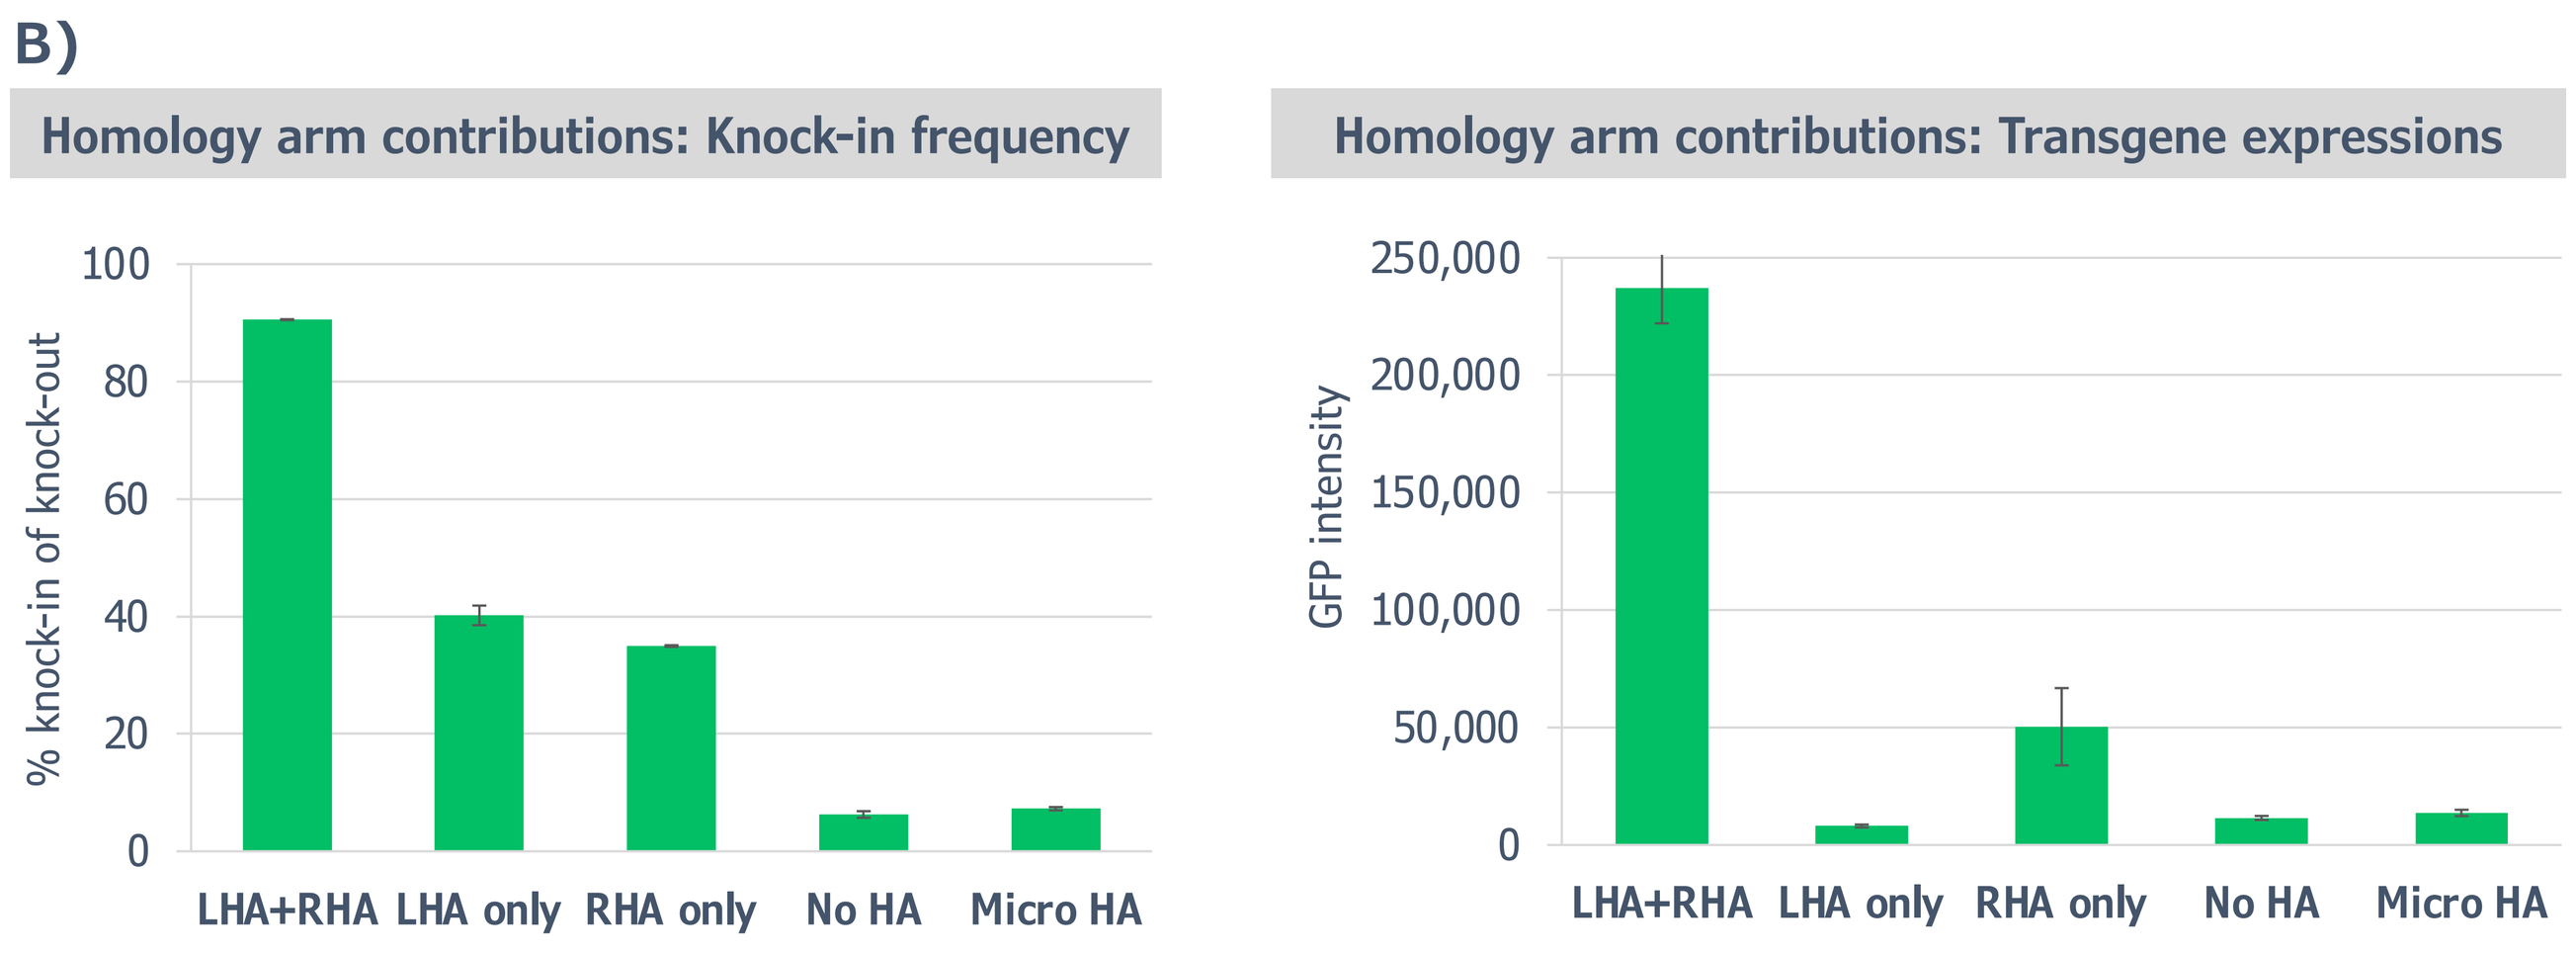
Homology arm contributions: Knock-in frequency
% knock-in of knock-out
Homology arm contributions: Transgene expressions
GFP intensity
LHA+RHA
LHA only
RHA only
No HA
Micro HA

C)

Description	Insert %	Insert-positive alleles			
		HDR-HDR	HDR-NHEJ	NHEJ-HDR	NHEJ-NHEJ
Full homology	88.90%	93%	3%	4%	0%
No homology	5.90%	NA	NA	NA	NA
LHA only	40.30%	3%	95%	1%	1%
RHA only	33.60%	12%	1%	86%	1%
Microhomology	7.00%	NA	NA	NA	NA

Figure 4: A staggered DSB with 3′ overhangs potentiates efficient HDR.

A) T lymphocytes were edited with ARCUS (3' 4bp staggered cut), ARCUS-TREX1 fusion (TREX1 removes 3' 4bp ends), or CRISPR-Cas9 (blunt cut) to generate the DSBs at the TRAC locus. **B)** Cells were transduced with HDR template across a range of multiplicities of infection (MOIs). Insertion frequency was significantly higher in the presence of a staggered DSB. **C)** Integration into the various DSBs was assessed following HDR inhibition (using trifluridine or B02), NHEJ inhibition (using NU7026), or MMEJ inhibition (using rucaparib). Only HDR inhibitors reduced integration into ARCUS DSBs. Inhibition of end-joining enhanced integration into blunt-ended DSBs suggesting the predominant repair mechanism for each.

A)

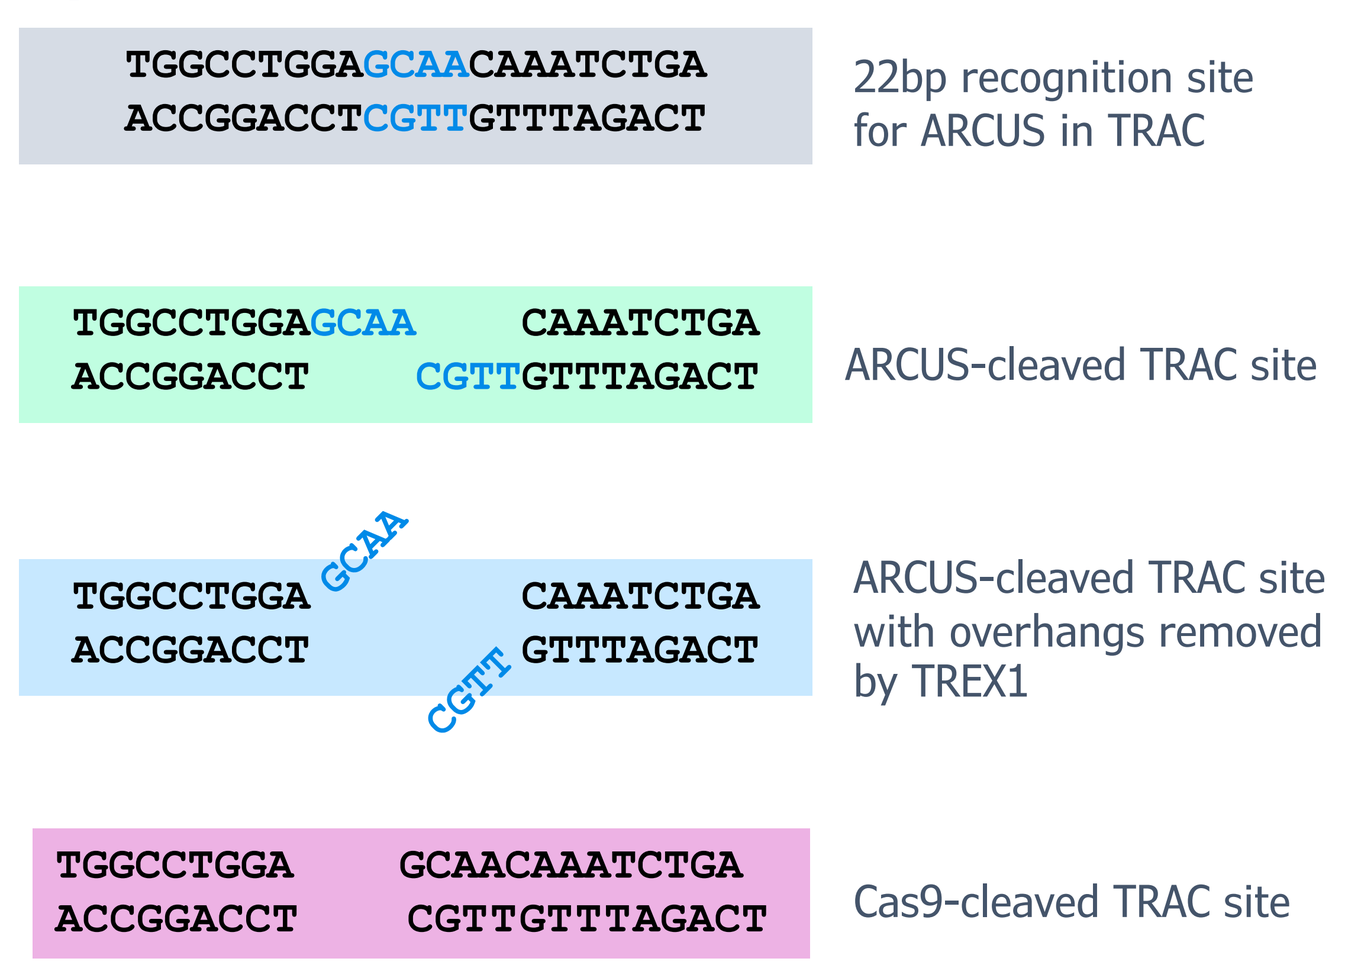
TGGCCTGGAGCAACAAATCTGA
ACCGGACCTCGTTGTTTAGACT
22bp recognition site for ARCUS in TRAC
TGGCCTGGAGCAA CAAATCTGA
ACCGGACCT CGTTGTTTAGACT
ARCUS-cleaved TRAC site
TGGCCTGGA GCAA CAAATCTGA
ACCGGACCT CGTT GTTTAGACT
ARCUS-cleaved TRAC site with overhangs removed by TREX1
TGGCCTGGA GCAACAAATCTGA
ACCGGACCT CGTTGTTTAGACT
Cas9-cleaved TRAC site

B)

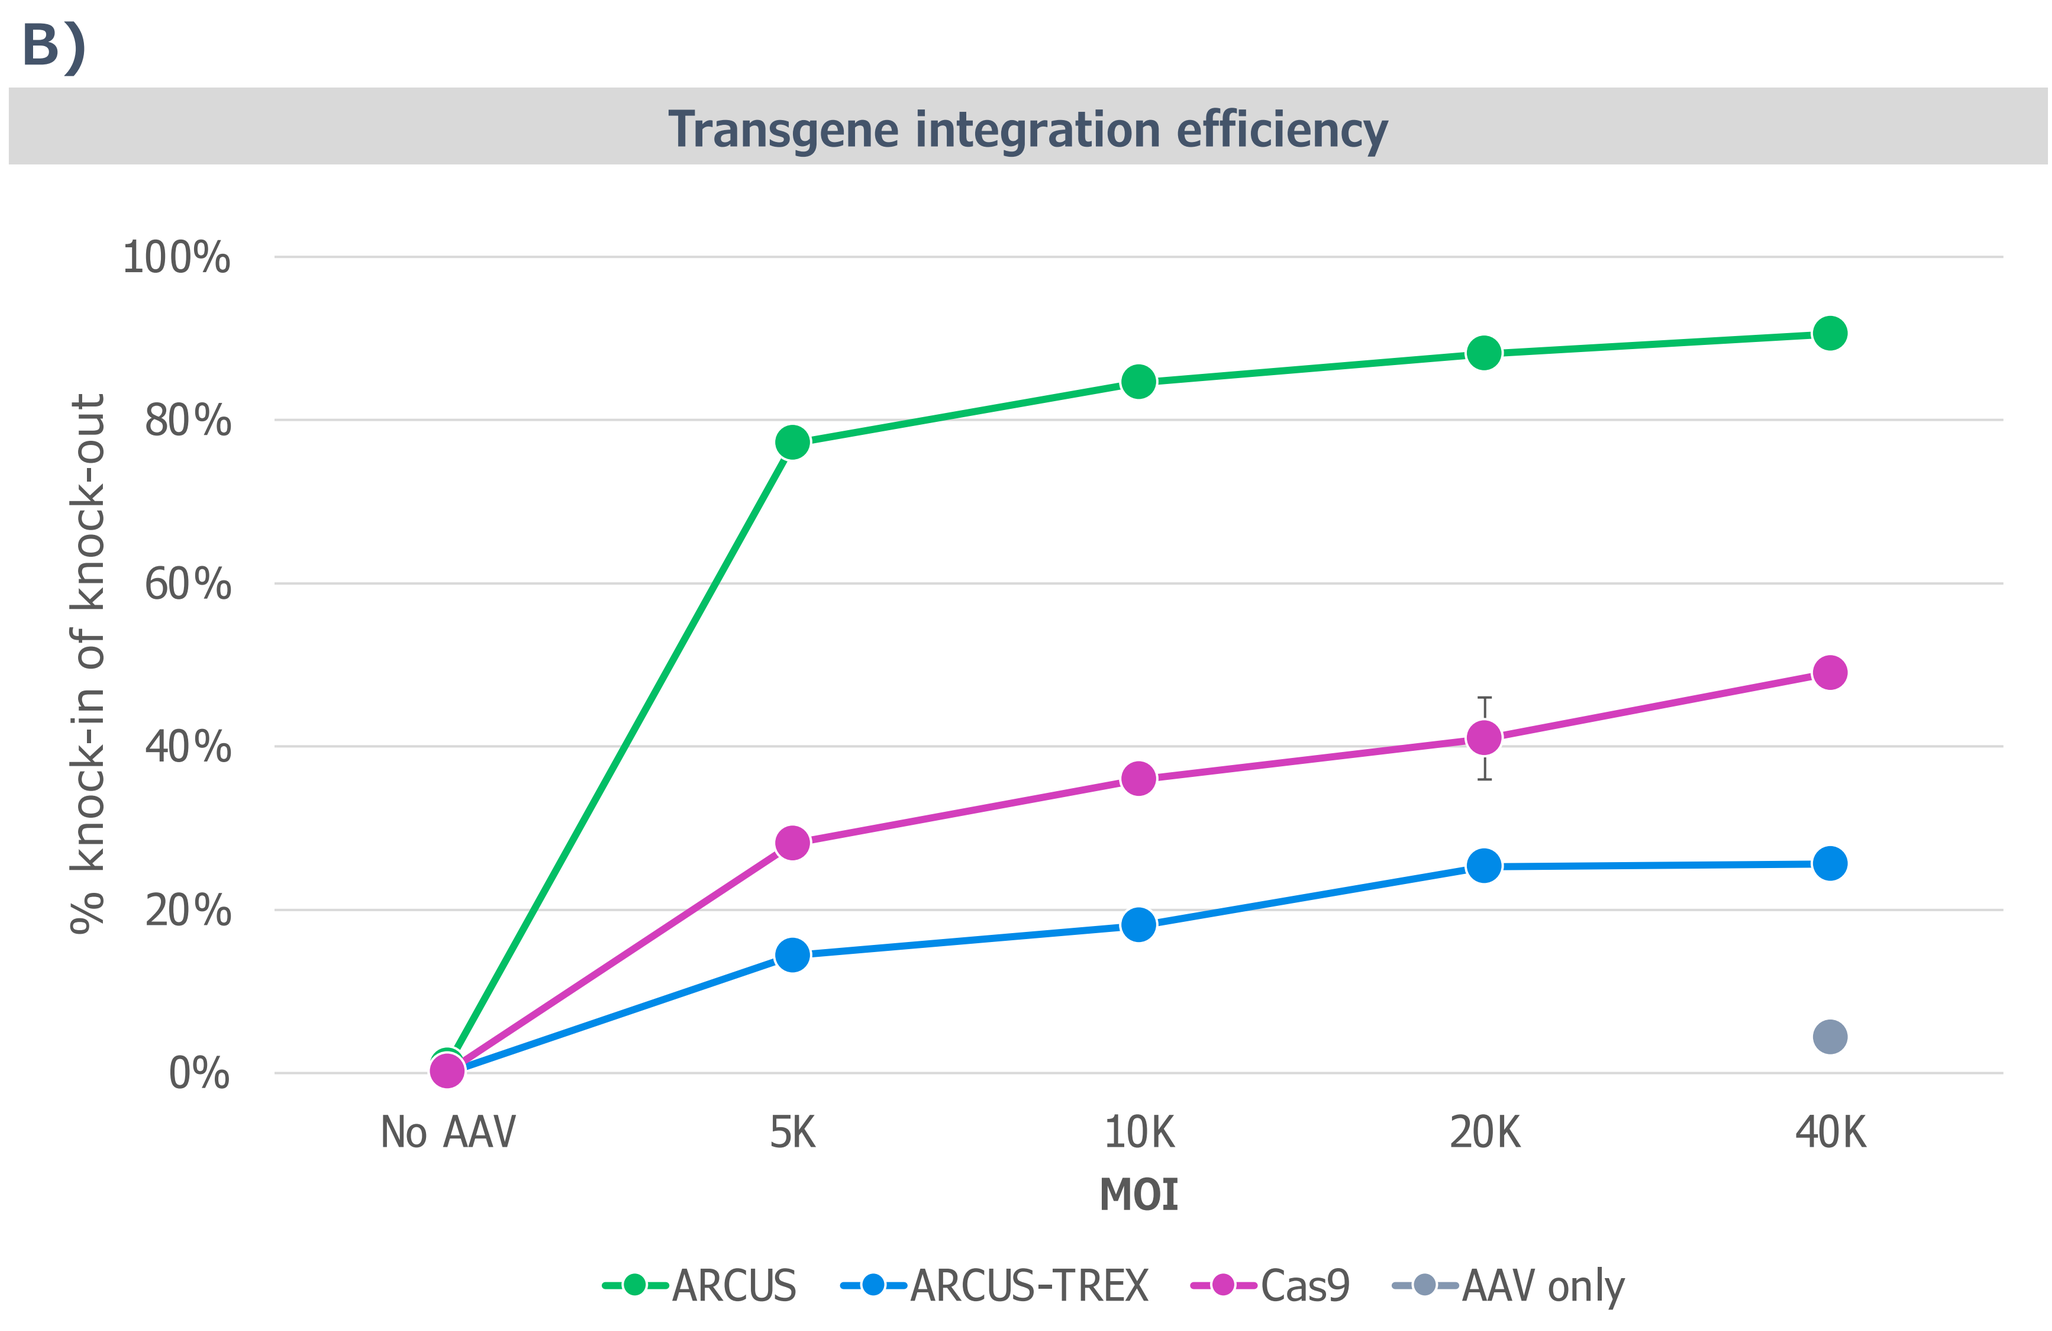
Transgene integration efficiency
% knock-in of knock-out
No AAV
5K
10K
20K
40K
MOI
ARCUS
ARCUS-TREX
Cas9
AAV only

C)

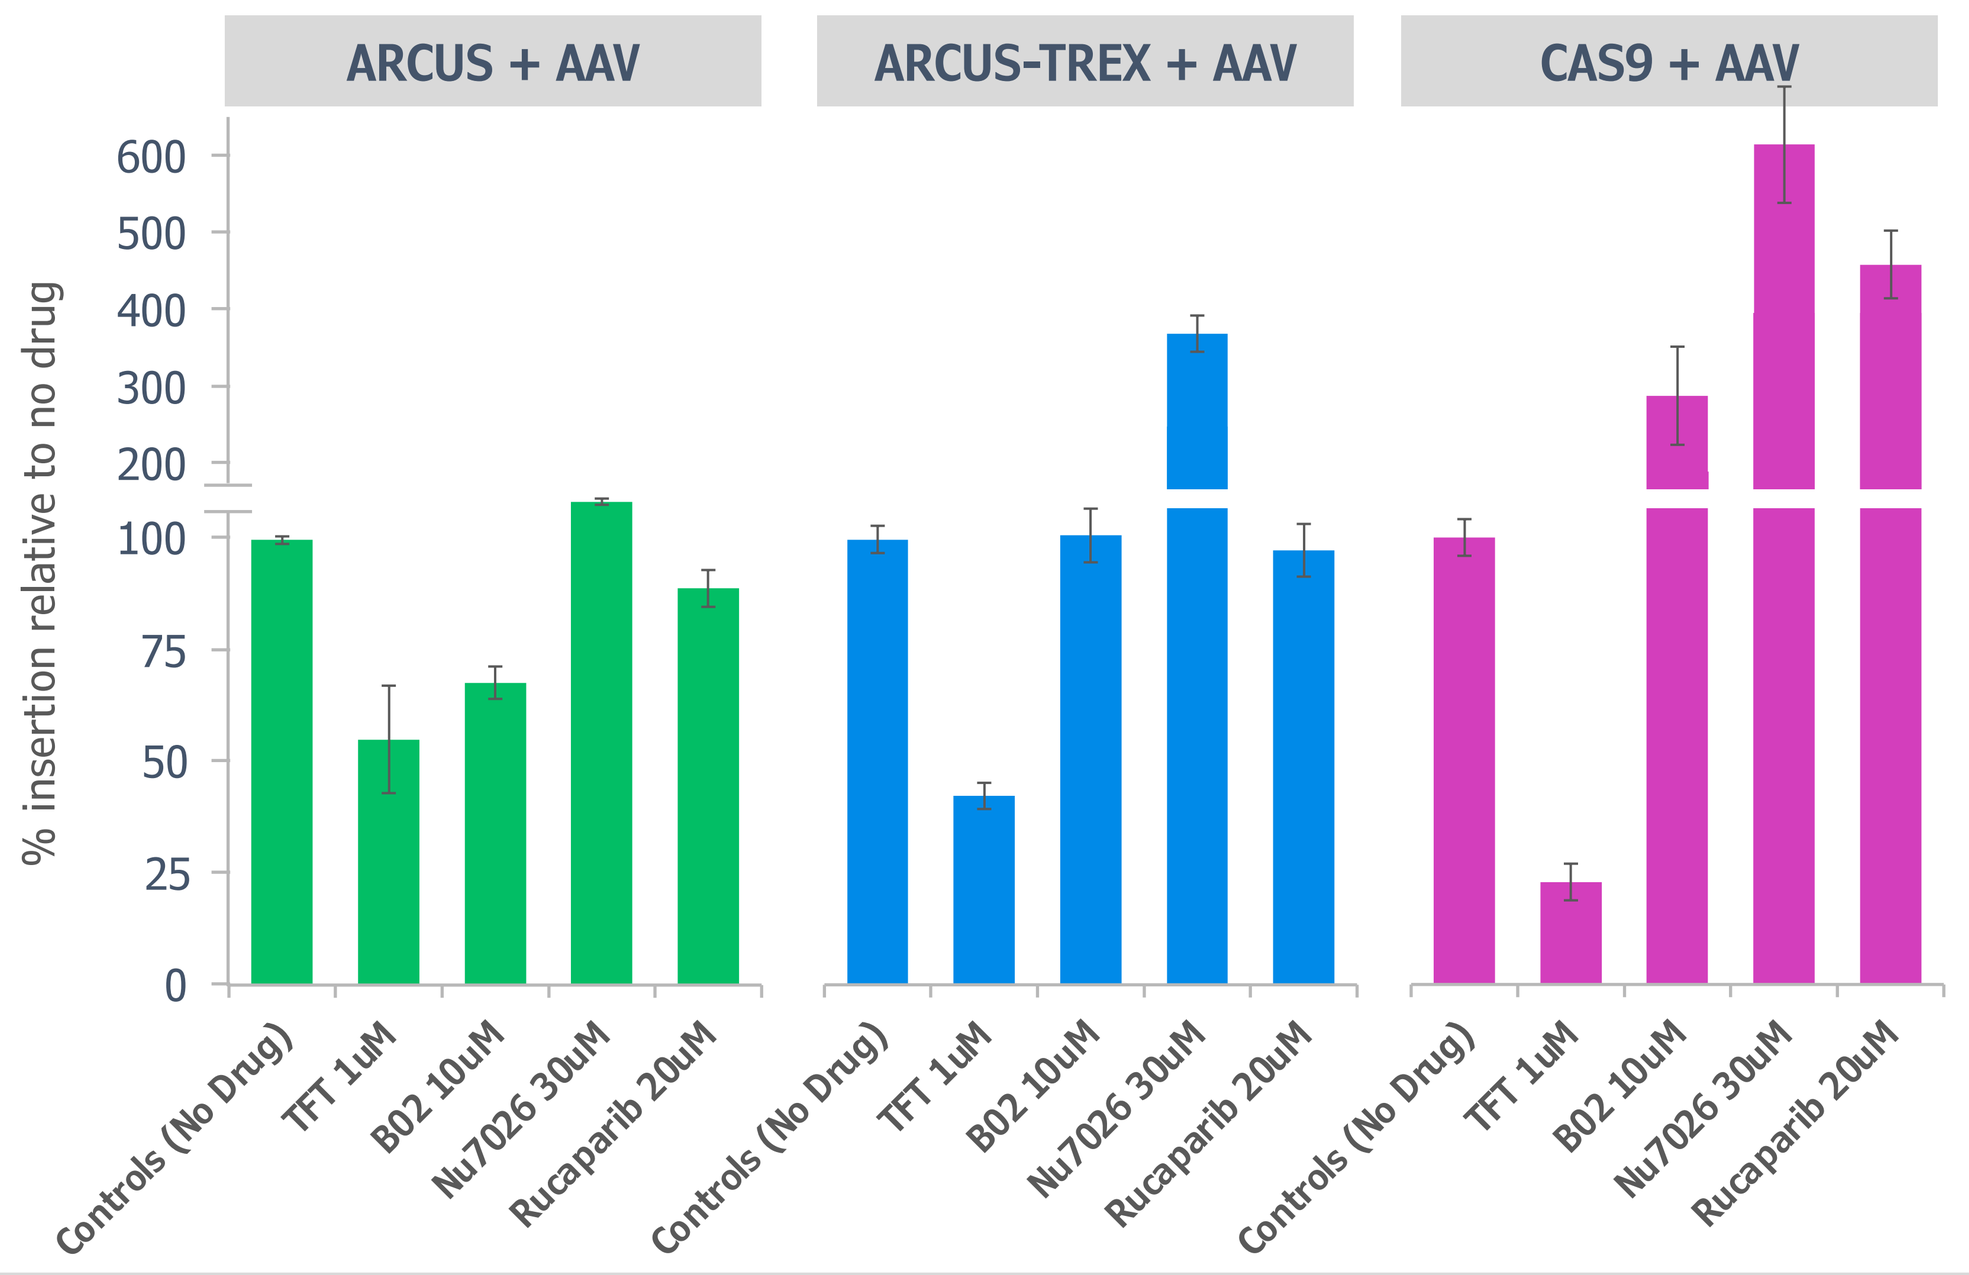
ARCUS + AAV
ARCUS-TREX + AAV
CAS9 + AAV
% insertion relative to no drug
Controls (No Drug)
TFT 1uM
B02 10uM
Nu7026 30uM
Rucaparib 20uM

Efficient HDR enables base changing, defined insertions & deletions, and gene replacement.

Figure 5: Mismatches in homology arms can change bases and codons.

A) T cells were edited with an ARCUS nuclease specific for the 5′UTR of TGFBR2, and the right homology arms were designed to alter the site of translation initiation . **B)** Single bp alterations, or 3bp insertions or deletions disrupted TGFBRII surface display with roughly equal frequencies . **C)** Additionally, AAV repair vectors were produced with degeneracy at each base in the start site, or an adjacent C. NGS found that bases were changed from wild-type in 39-47% of reads. Of those reads, each base could be changed to any other base with approximately equal frequency, indicating base editing via HDR is capable of small insertions, deletions, and all possible base changes.

A)

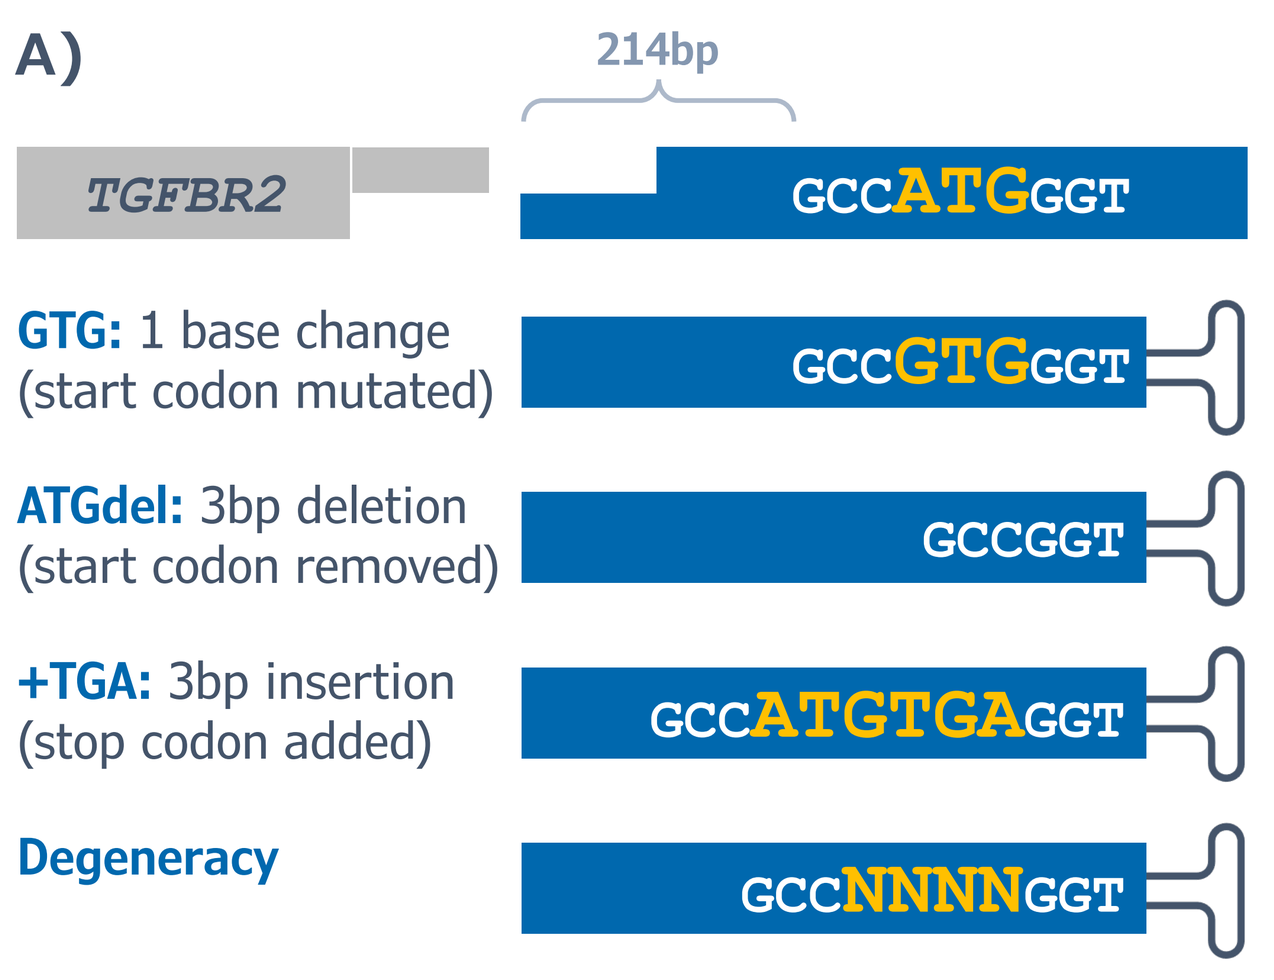
214bp
TGFBR2
GCCATGGGT
GTG: 1 base change
(start codon mutated)
GCCGTGGGT
ATGdel: 3bp deletion
(start codon removed)
GCCGGT
+TGA: 3bp insertion
(stop codon added)
GCCATGTGAGGT
Degeneracy
GCCNNNNGGT

B)

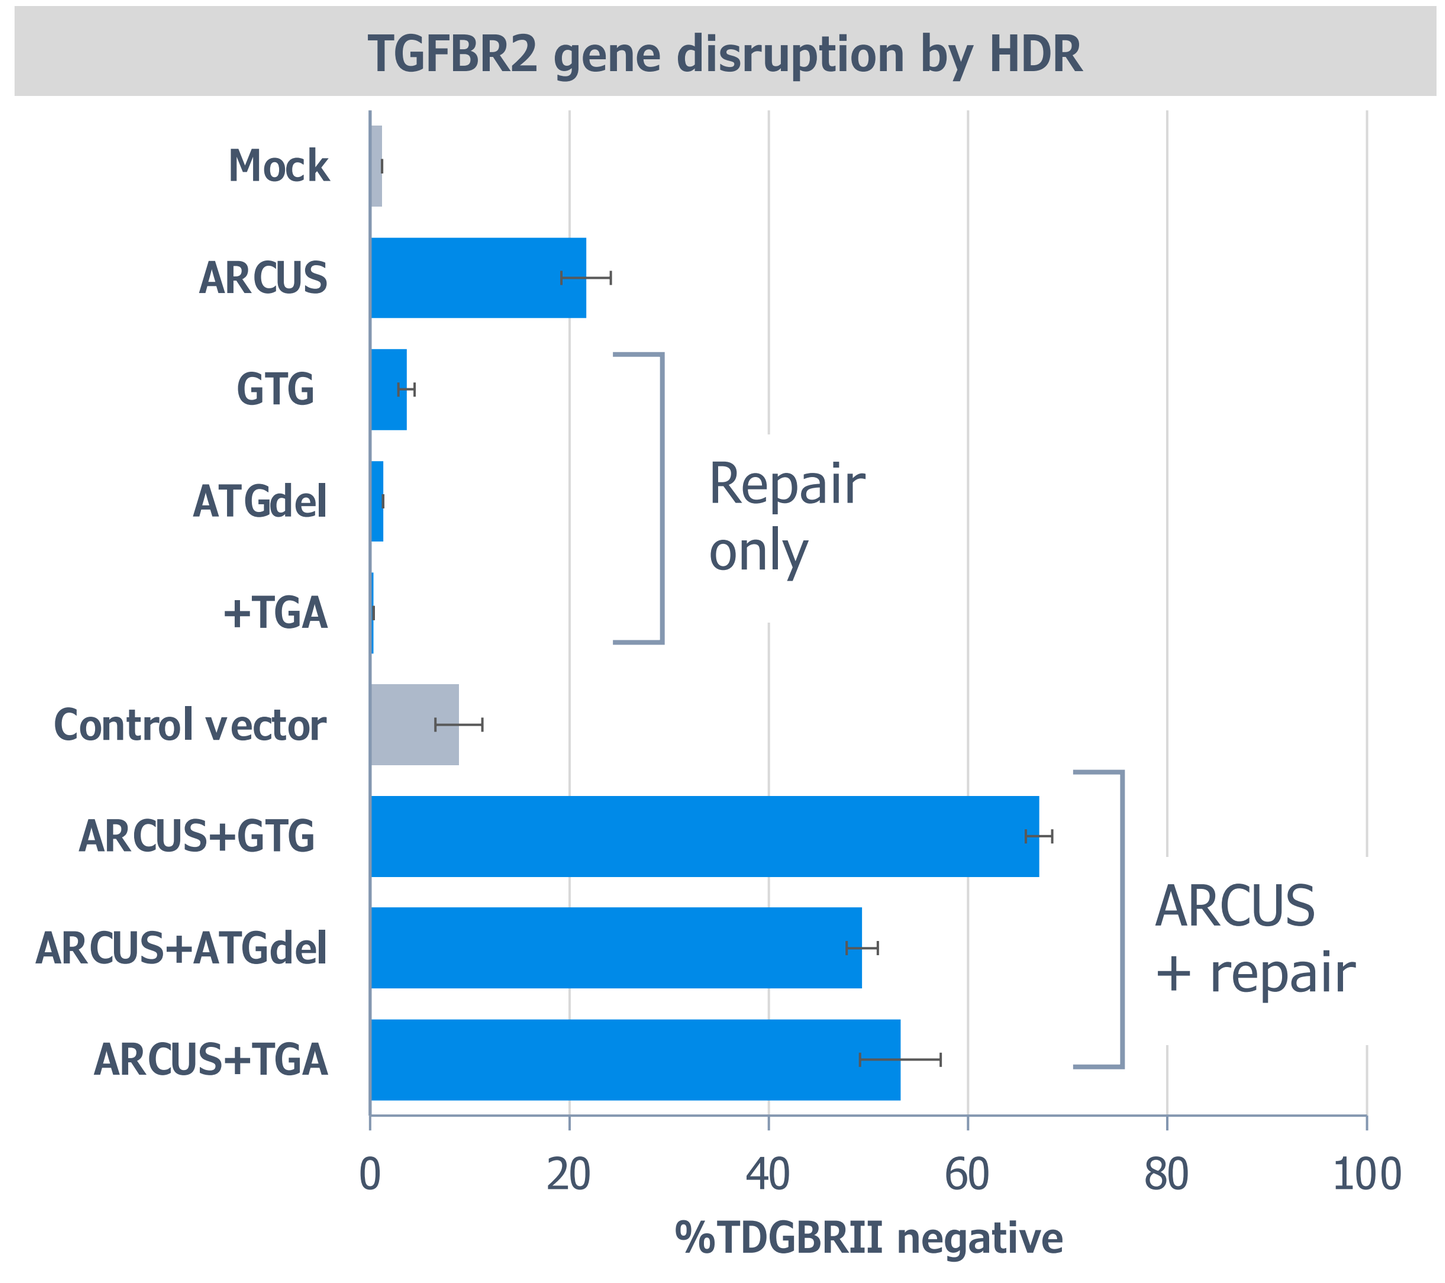
TGFBR2 gene disruption by HDR
Mock
ARCUS
GTG
ATGdel
+TGA
Control vector
ARCUS+GTG
ARCUS+ATGdel
ARCUS+TGA
Repair only
ARCUS + repair
%TDGBRII negative

C)

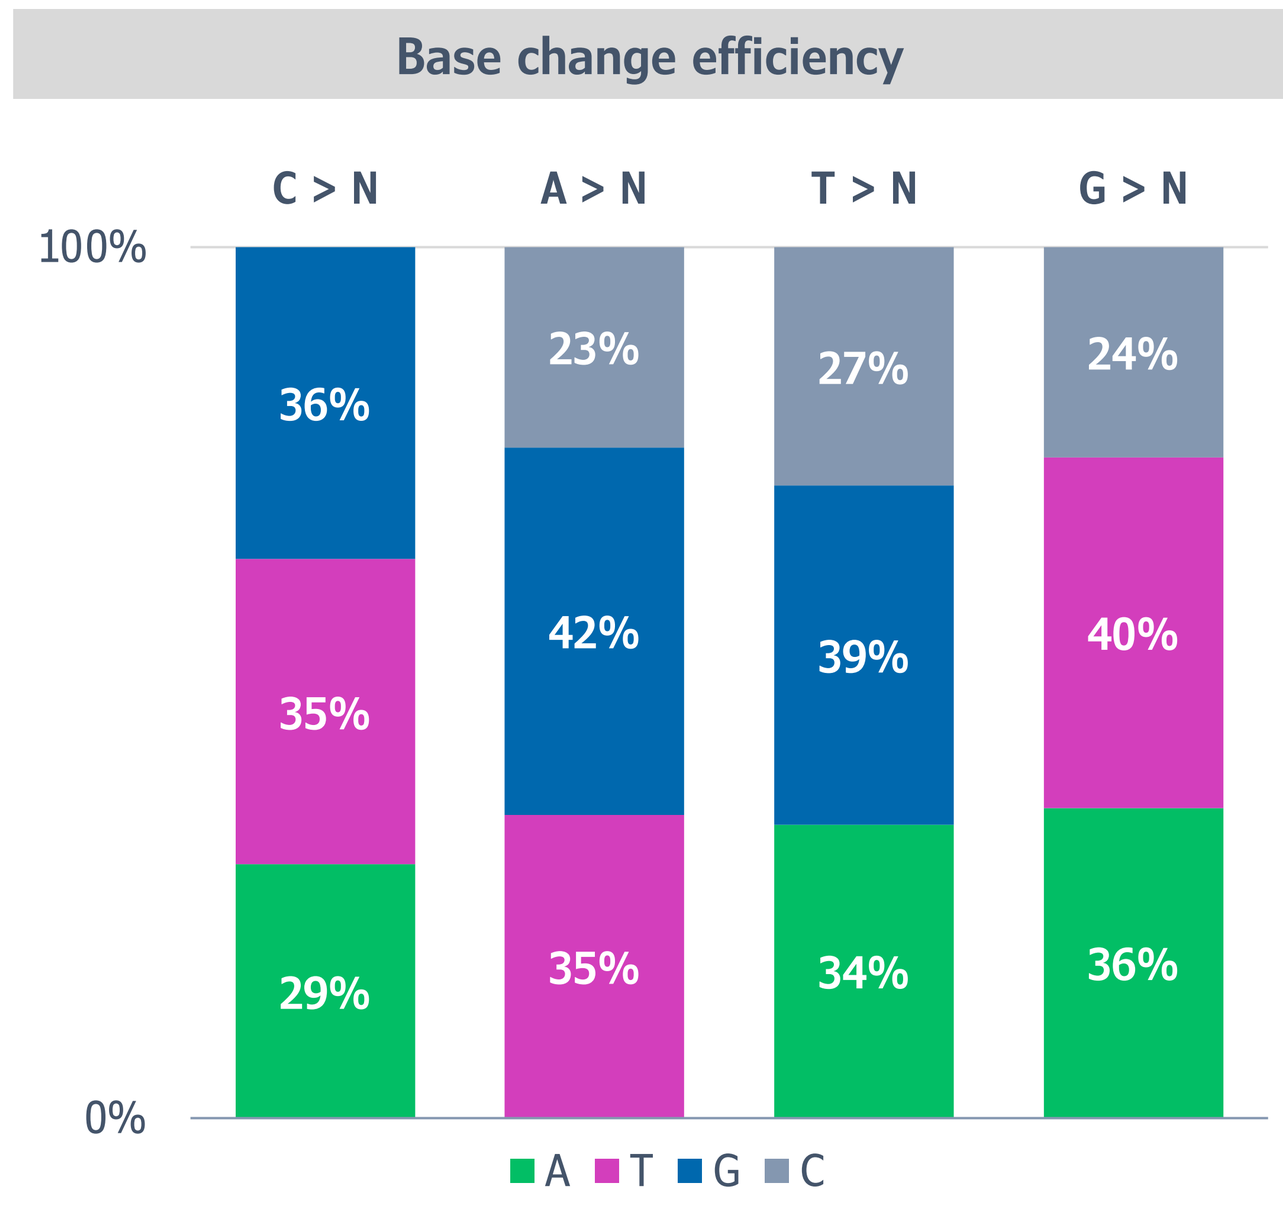
Base change efficiency
C > N
A > N
T > N
G > N
36%
35%
29%
23%
42%
35%
27%
39%
34%
24%
40%
36%
A
T
G
C

Figure 6: Genomic regions can be replaced by priming HDR at DSB-distal sites.

A) T cells were edited with a TRAC-specific nuclease and the locus was repaired with a template containing an LHA adjacent to the cut and a RHA 2.3kb downstream of the cut. HDR repair replaces the second and third exon and introns with an artificial exon 2/3 and an intron containing a B2M shRNAmir. **B)** Correct HDR repair reconstitutes TRAC and knocks down B2M. Flow cytometry identified populations of insert-bearing edited alleles, and sorted samples were subjected to Nanopore long-read sequencing. **C)** Nearly two thirds of cells exhibited the replaced allele, and over 90% of the reads suggested the repair was correct.

A)

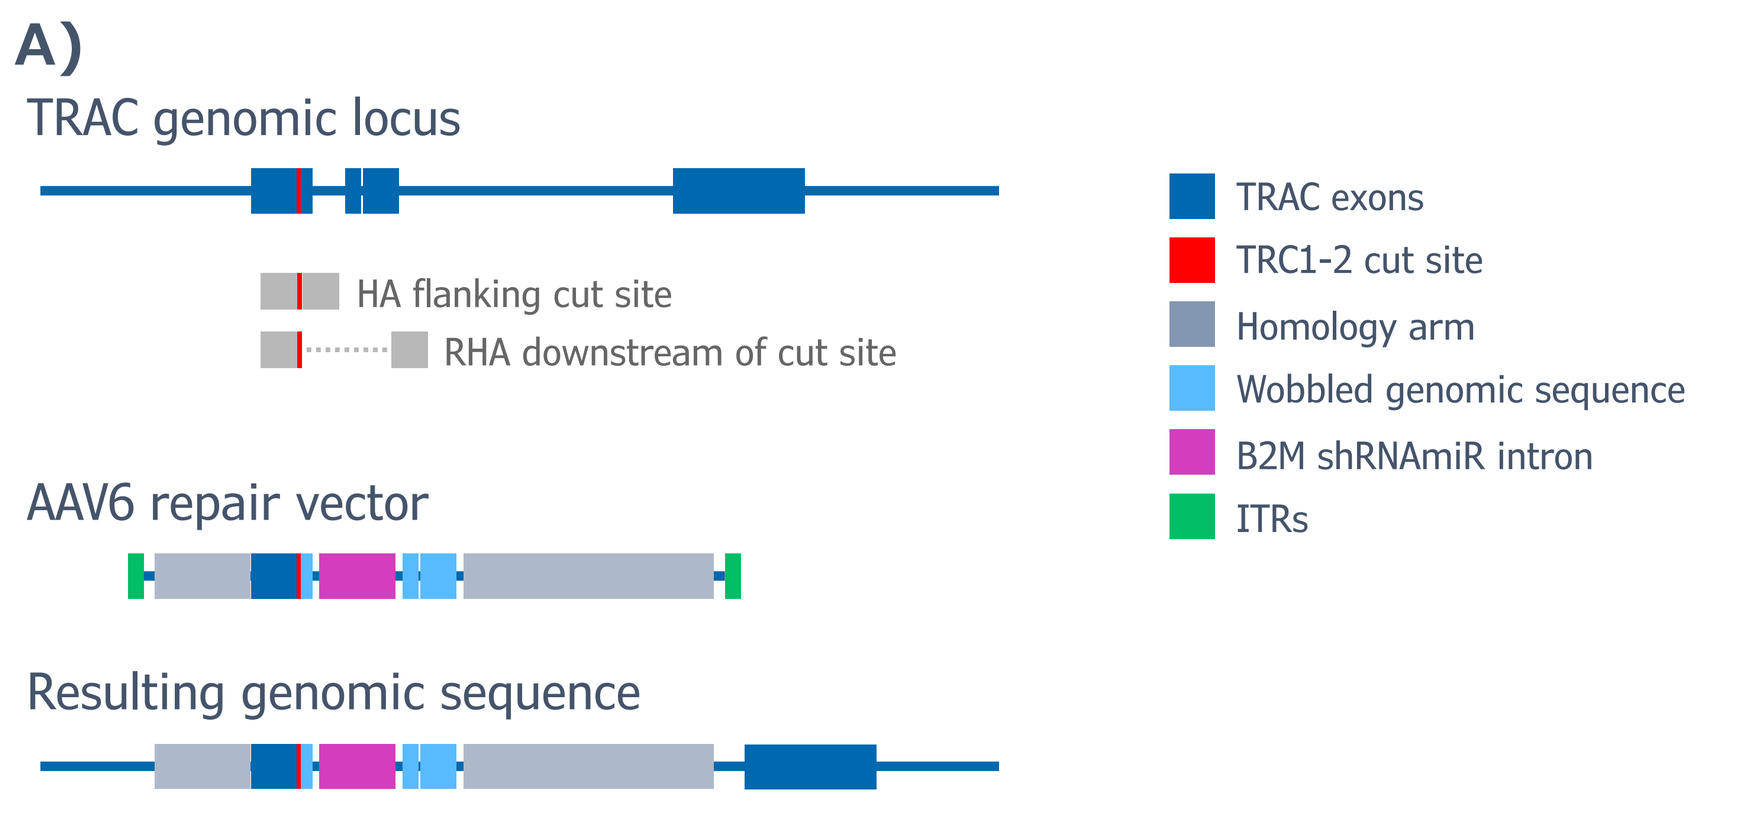
TRAC genomic locus
HA flanking cut site
RHA downstream of cut site
AAV6 repair vector
Resulting genomic sequence
TRAC exons
TRC1-2 cut site
Homology arm
Wobbled genomic sequence
B2M shRNAmiR intron
ITRs

B)

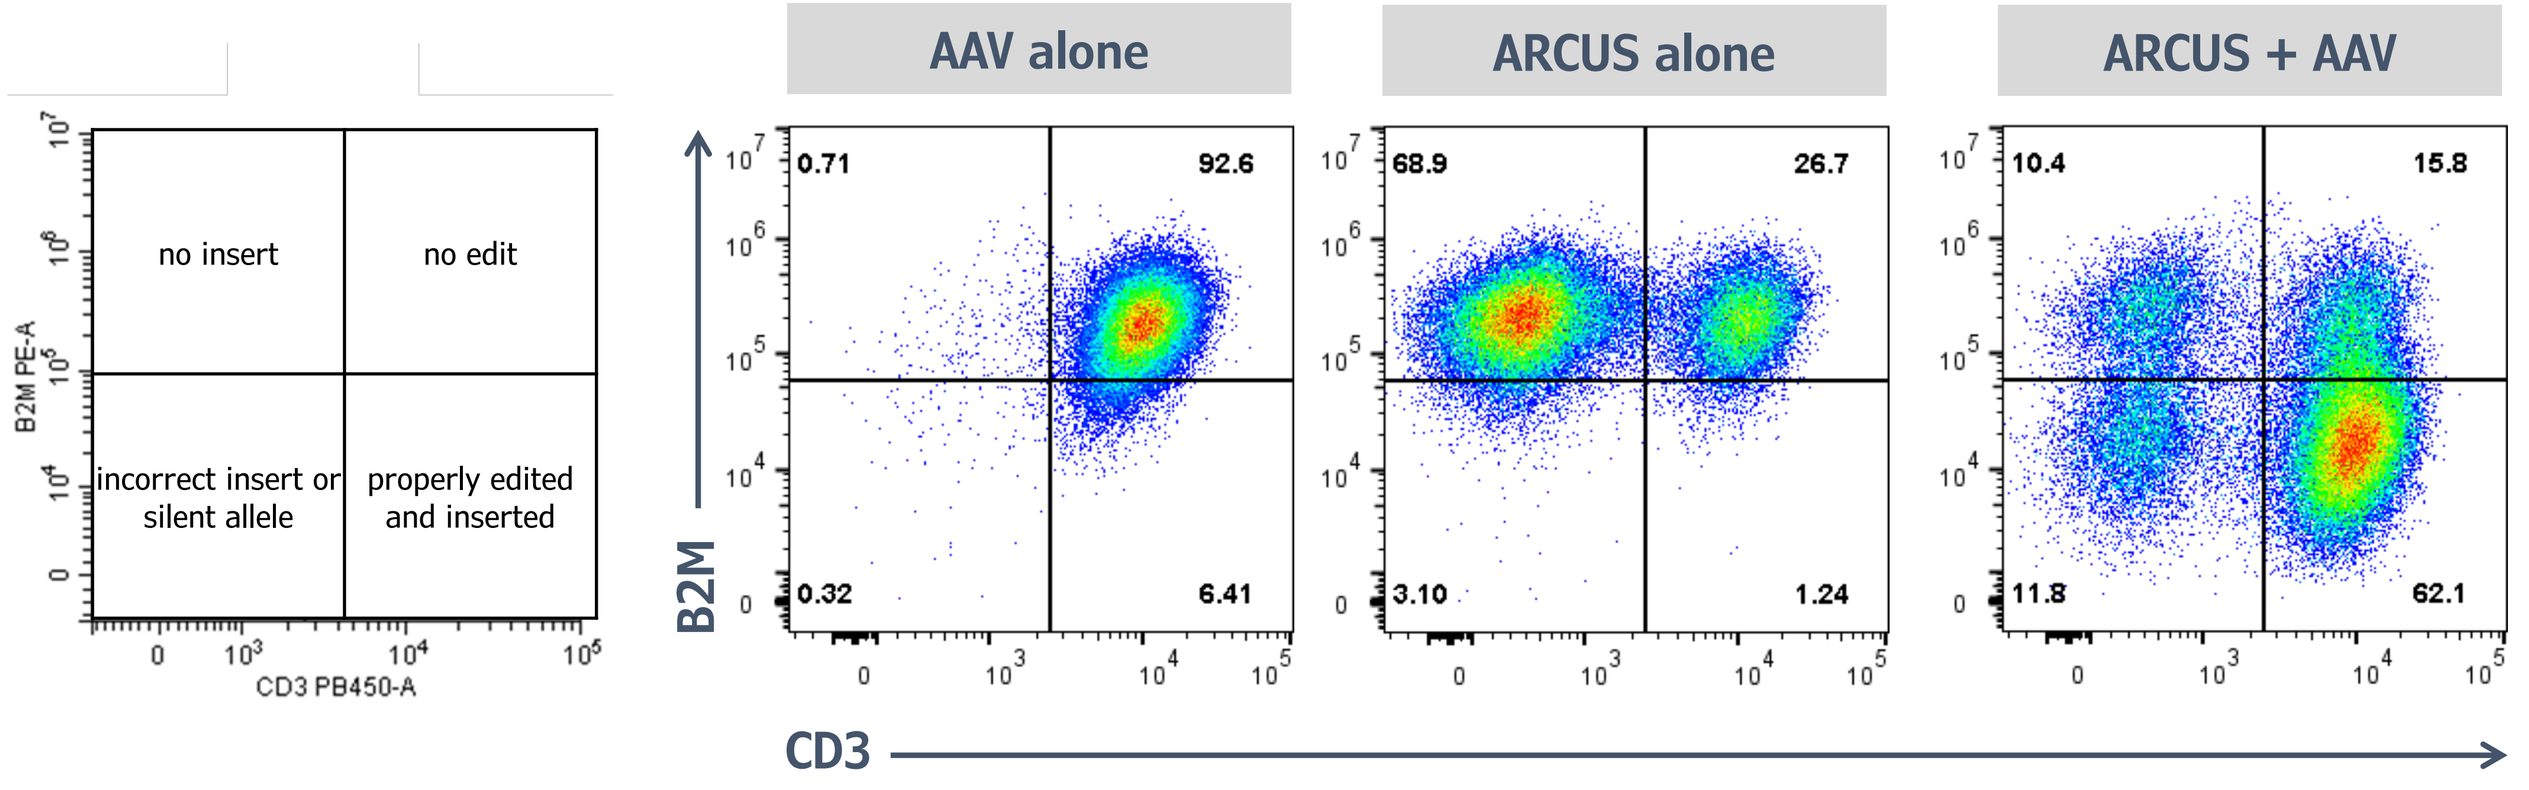
no insert
no edit
incorrect insert or silent allele
properly edited and inserted
AAV alone
ARCUS alone
ARCUS + AAV
0.71
92.6
0.32
6.41
68.9
26.7
3.10
1.24
10.4
15.8
11.8
62.1
B2M
CD3

C)

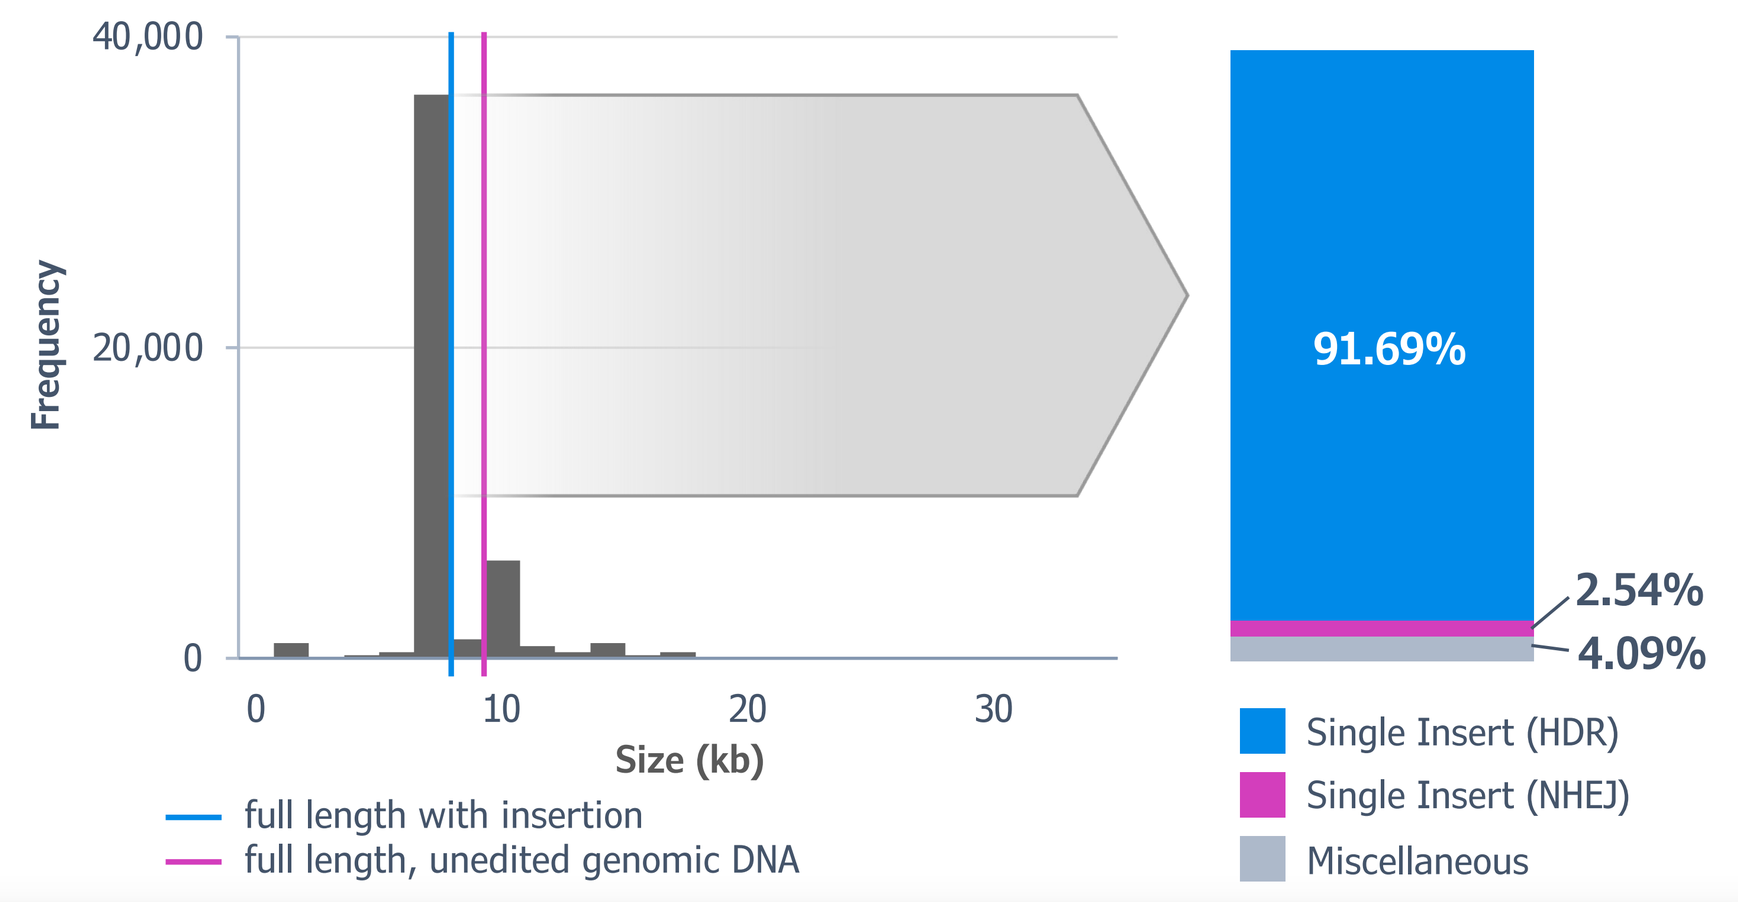
Frequency
Size (kb)
full length with insertion
full length, unedited genomic DNA
91.69%
2.54%
4.09%
Single Insert (HDR)
Single Insert (NHEJ)
Miscellaneous

CONCLUSIONS

- ARCUS drove a high rate of gene insertion into its cut site in both T-cells and noncycling PHH.
- Repair and gene insertion took place via HDR.
 - Efficient insertion required sufficient length homology arms on both sides of insert.
 - Sequence analysis did not detect ITR capture, repeated elements, or other indicators of NHEJ.
 - Pharmacological inhibitors of HDR reduced integration into ARCUS DSBs while end-joining inhibitors do not.
- The staggered cut made by ARCUS was required for efficient HDR; eliminating the overhang with an ARCUS-TREX1 fusion or using a blunt-cutting enzyme such as Cas9 significantly reduced HDR rates.
- Mismatches in a homology arm were tolerated and can be leveraged to change individual bases or codons. All 12 possible base changes were achieved with similar efficiency using ARCUS and a HDR repair template.
- Large stretches of genomic DNA were replaced by targeting a homology arm to a DSB-distal site.

PRECISION BIOSCIENCES

ESGCT Annual Congress. October 2024. Rome, Italy