3510

Optimization of targeted gene insertion using ARCUS nucleases

Wendy K. Shoop[1], Emma Sevigny[1], Paige Nemec[1], Janel Lape[1], Jeff Smith[1], Ying K. Tam[2], Paulo J.C. Lin[2], Sean C. Semple[2], Derek Jantz[1], Cassandra L. Gorsuch[1]

[1]Precision BioSciences, Durham, NC, USA; [2]Acuitas Therapeutics, Vancouver, British Columbia, Canada

INTRODUCTION

- Replacement of missing or mutated genes *in vivo* via adeno associated virus (AAV) gene therapy has shown promising clinical results in numerous indications.
- Gene expression from episomal AAV is hampered by vector dilution in tissues that experience cellular turnover, potentially limiting the therapeutic durability.
- Nuclease-driven, targeted gene insertion is an appealing therapeutic approach to overcome these challenges.
- By inserting a wild-type version of a mutated DNA sequence into the genome of affected cells, functional protein can be produced, and edits can be permanently retained through cell division and potentially lead to longer durability of therapeutic effect.
- ARCUS is a single-component protein derived from I-*Cre*I which contains both a 22 bp site-specific DNA recognition interface and endonuclease activity.
- When ARCUS nucleases engage their intended target site, a double-strand break occurs, resulting in a 4-base pair (bp), 3' overhang.
- We hypothesized that the ARCUS-induced 3' overhang may facilitate high levels of gene insertion.
- Here we demonstrate an improved ability to identify and optimize ARCUS nucleases specifically for gene insertion. We further explored increasing insertion frequency through modifications of the gene insertion AAV construct.

METHODS

- Immortalized HEK 293 cells were nucleofected either with ARCUS mRNA alone, or ARCUS mRNA in conjunction with a dsDNA repair template. The repair template contained 300 bp of homology to the sequence surrounding the cut site.
- Indels at the target site were quantified using digital PCR (dPCR).
- Non-dividing primary human hepatocytes (PHHs) were co-transfected with ARCUS, either delivered by AAV or LNP (Acuitas Therapeutics), and a repair template delivered by AAV. The repair template contained 300 bp of homology to the sequence surrounding the cut site.
- Genomic DNA was collected from both cell types and evaluated for productive and non-productive insertion events using dPCR.
- Two different dPCR assays were used to differentiate productive insertions from non-productive insertions. An insertion event which occurs in the forward orientation is considered productive, whereas one which occurs in the reverse orientation is considered non-productive. Forward-oriented insertion events can occur due to homology-directed repair (HDR, **Figure 1A**) or non-homologous end joining (NHEJ, **Figure 1B**), and these two types of repair can not be distinguished by the assay. Reverse-oriented insertion events can only occur due to NHEJ (**Figure 1C**).
- We assume that the frequency of NHEJ in the reverse orientation is equivalent to the frequency of NHEJ in the forward orientation, thus allowing us to calculate the relative contributions of HDR and NHEJ to the repair event.

RESULTS

Figure 2. ARCUS nucleases generate high levels of targeted gene disruption in HEK 293s

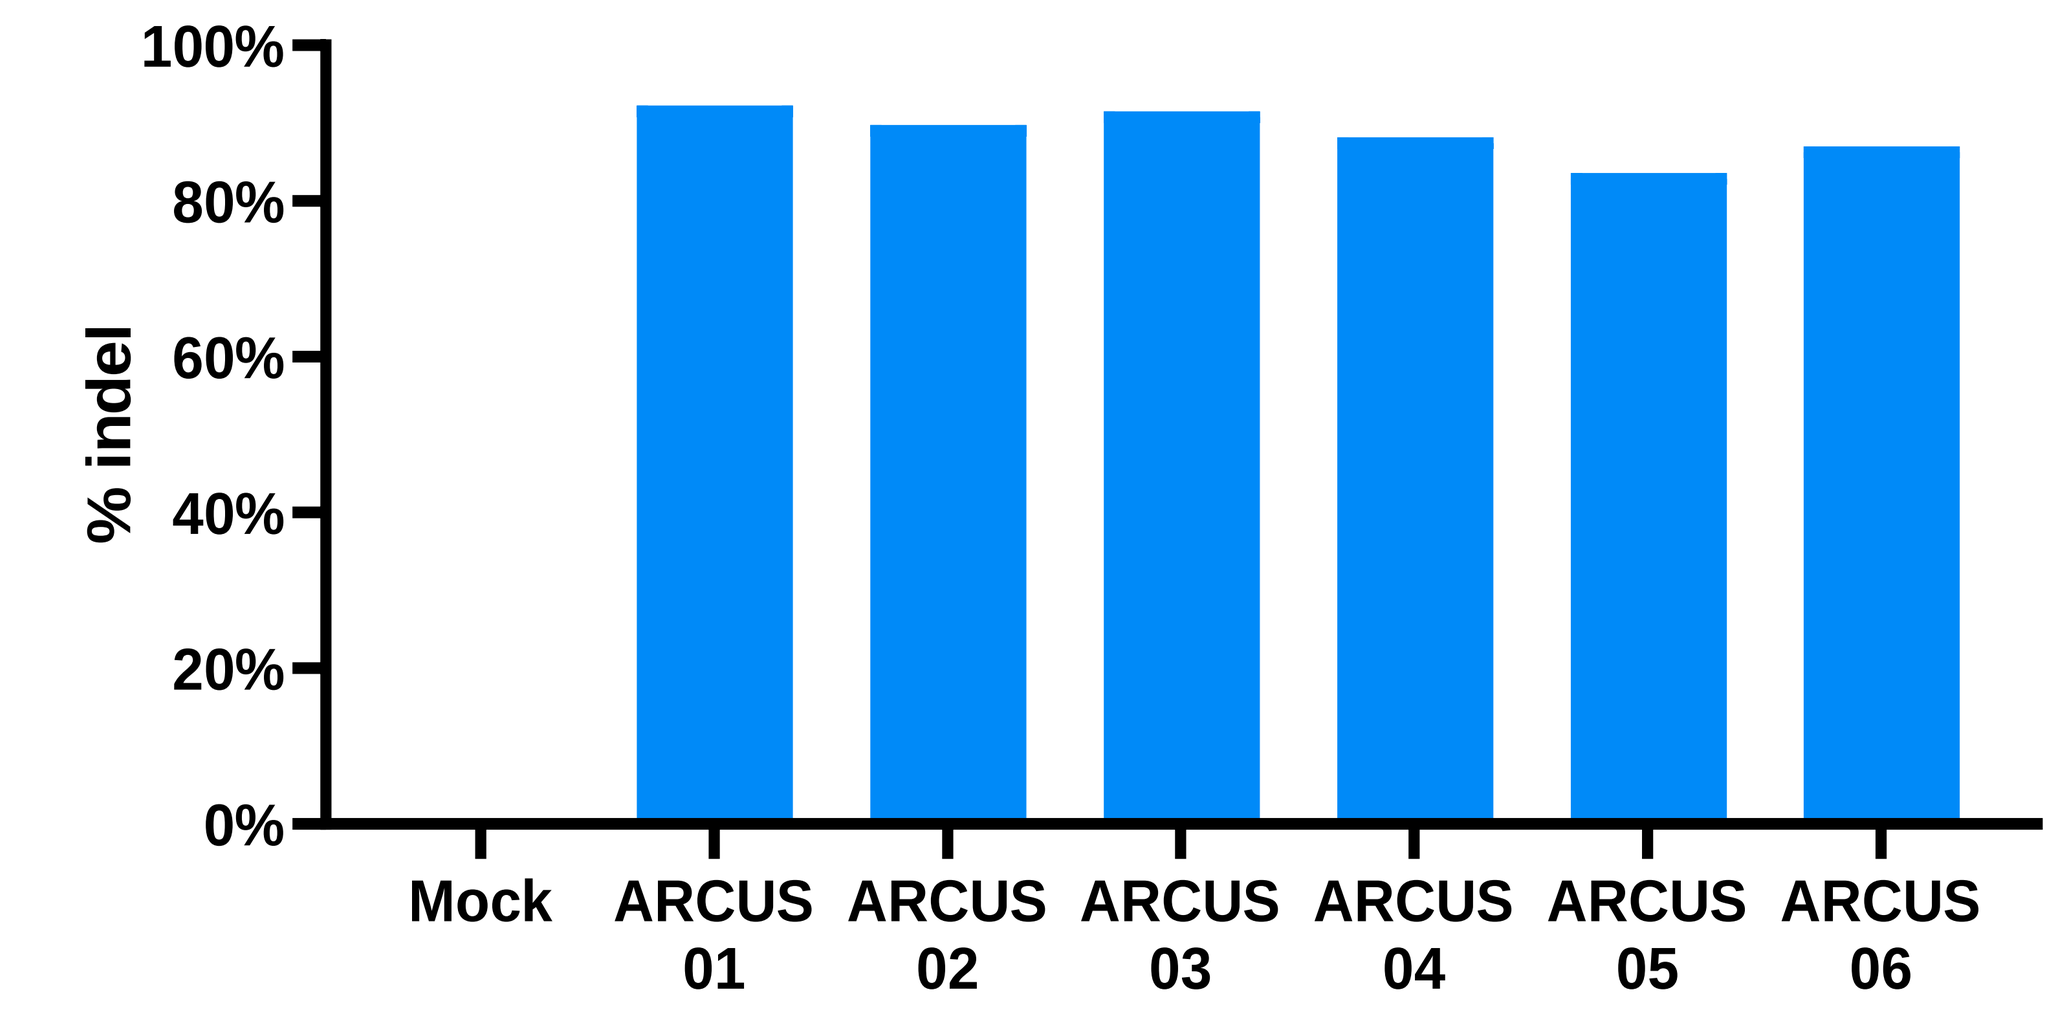
% indel
100%
80%
60%
40%
20%
0%
Mock
ARCUS 01
ARCUS 02
ARCUS 03
ARCUS 04
ARCUS 05
ARCUS 06

- ARCUS nucleases were evaluated in HEK 293 cells for indel formation at their intended target site. High levels of on-target activity (>88% indel formation) were demonstrated by all nucleases (**Figure 2**). ARCUS01 was the most potent, generating 92.3% indels at the cut site.

Figure 3. ARCUS01, when co-delivered with a repair template, produces 4-6% productive targeted insertions in HEK 293s and PHHs

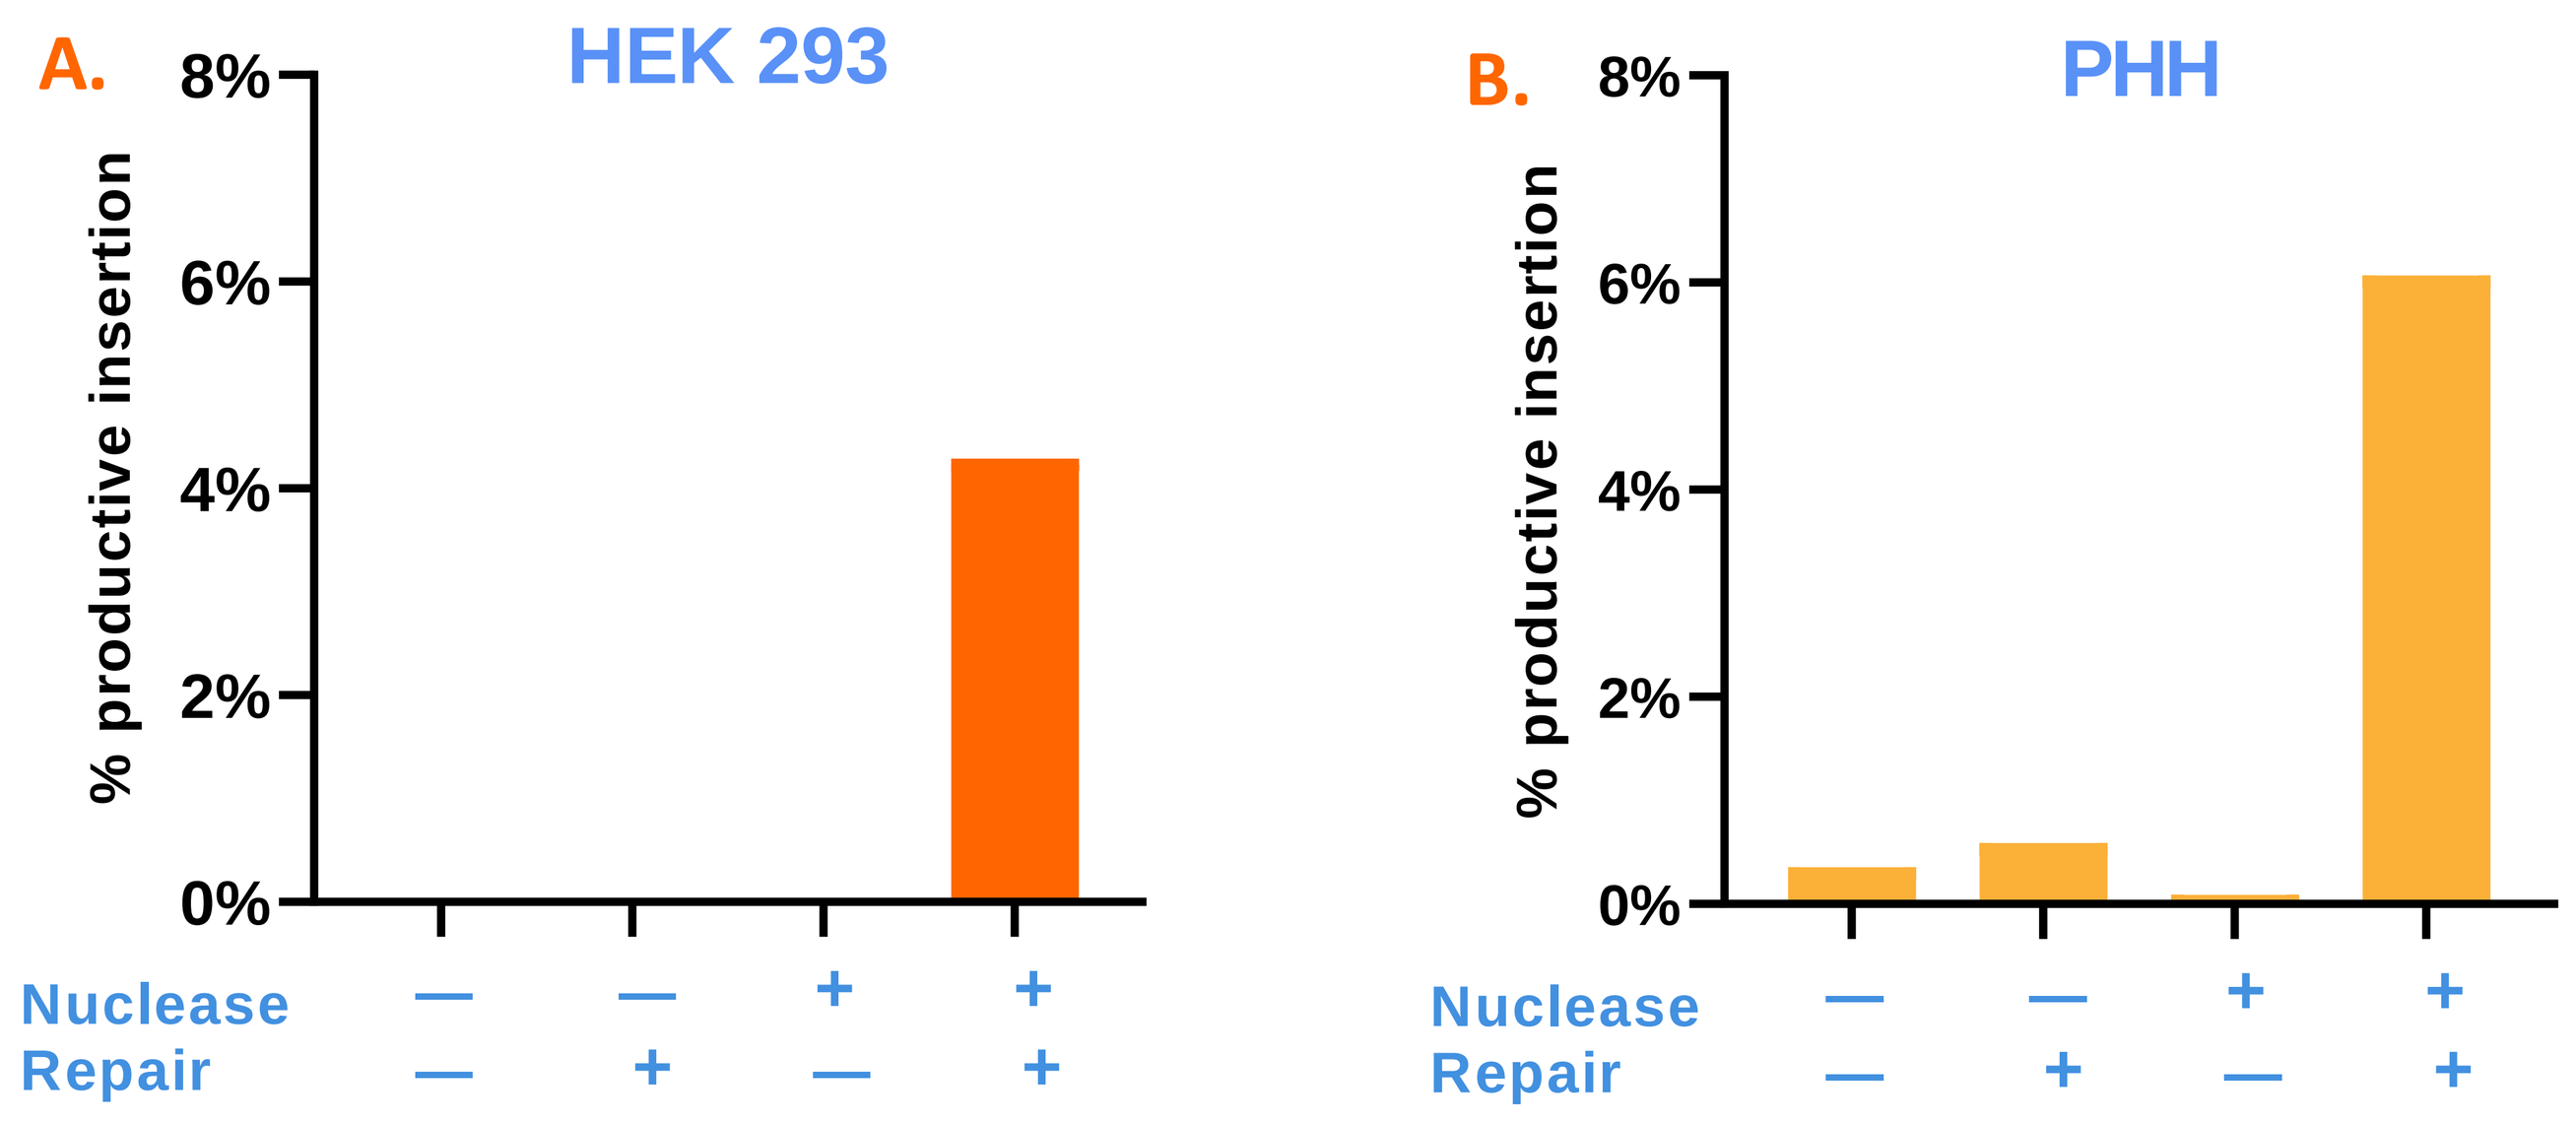
A.
HEK 293
% productive insertion
8%
6%
4%
2%
0%
Nuclease — — + +
Repair — + — +
B.
PHH
% productive insertion
8%
6%
4%
2%
0%
Nuclease — — + +
Repair — + — +

- ARCUS01 mRNA was co-transfected into HEK 293s with a dsDNA repair template. 4.29% productive insertions were generated at the cut site (**Figure 3A**).
- ARCUS01 was co-transfected into PHHs with a repair template, both delivered by AAV. A total of 6.07% productive insertions were quantified at the target site (**Figure 3B**), with 2.5% coming from HDR.

Figure 4. ARCUS nucleases display varying degrees of targeted insertion efficiency in HEK 293s

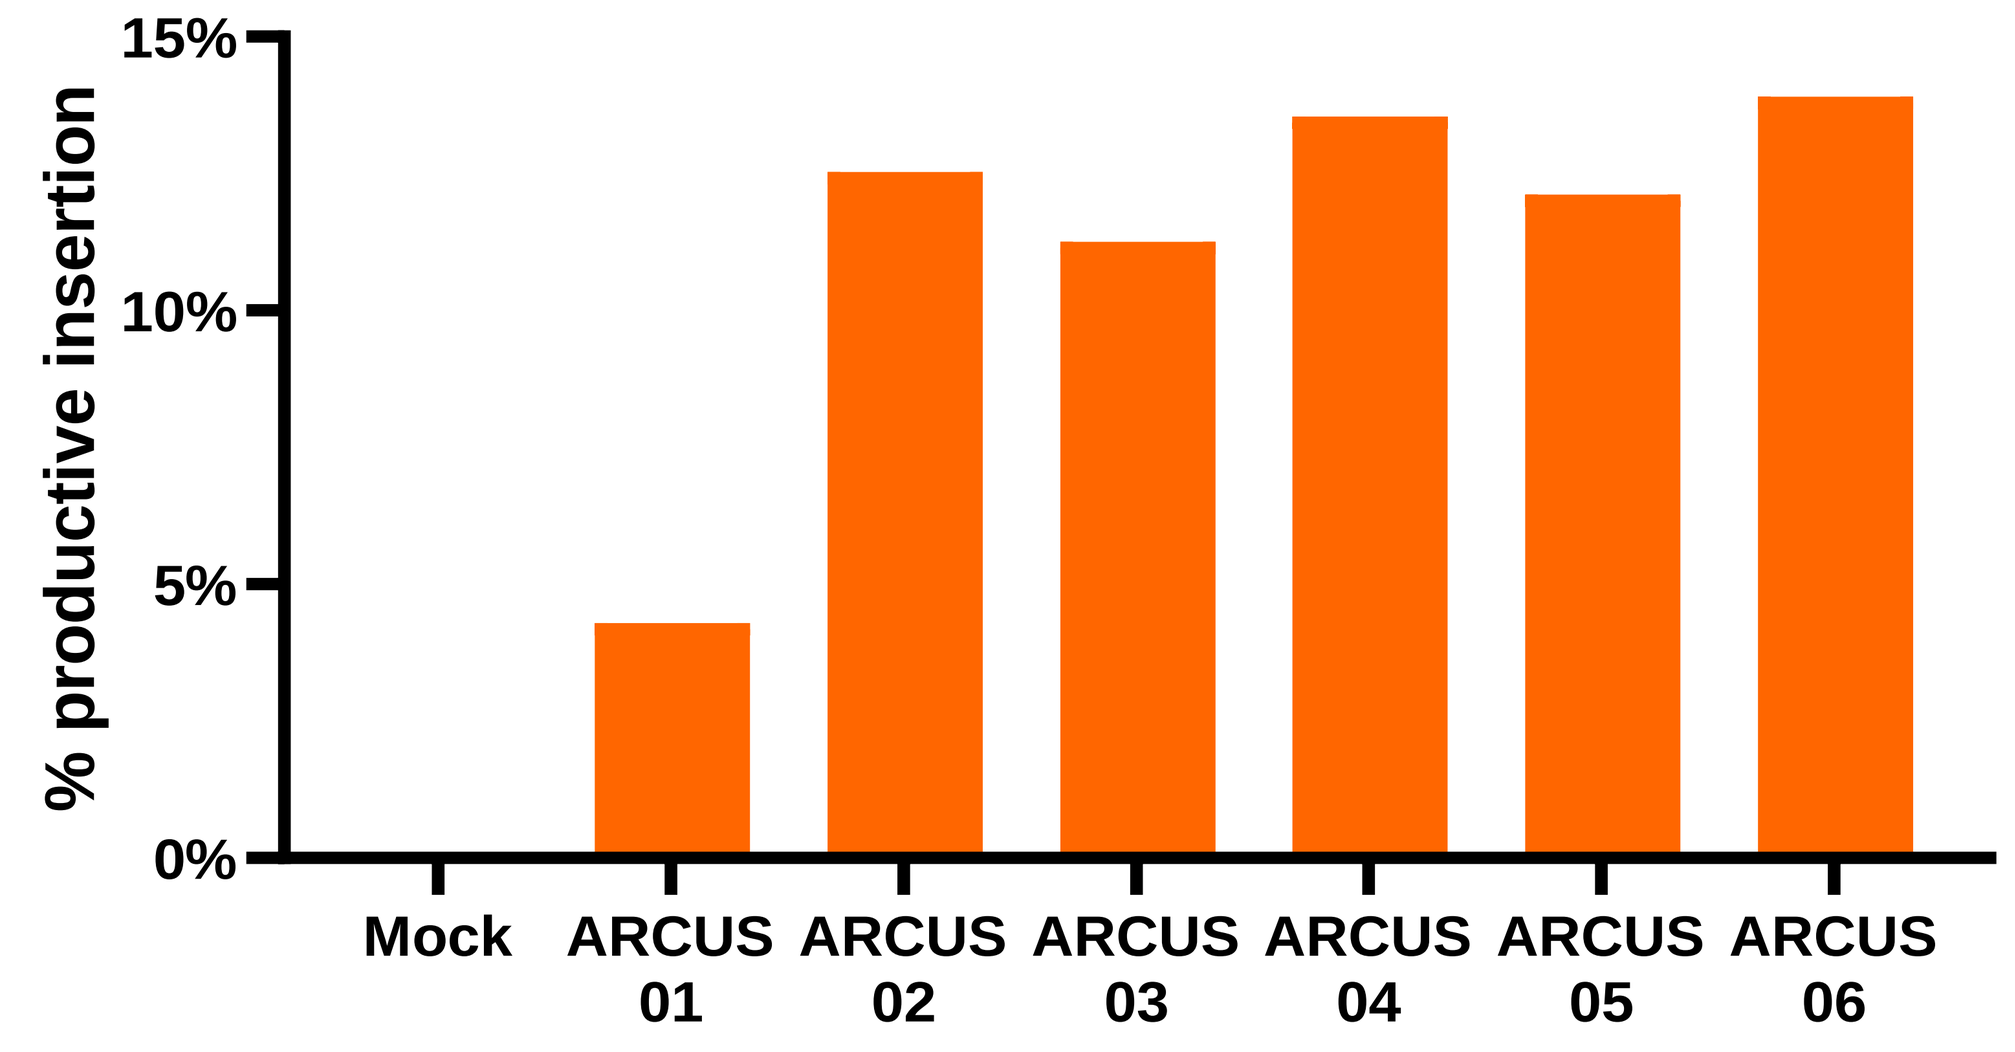
% productive insertion
15%
10%
5%
0%
Mock
ARCUS 01
ARCUS 02
ARCUS 03
ARCUS 04
ARCUS 05
ARCUS 06

- Based on the discrepancy between indels and targeted insertion observed with ARCUS01, we hypothesized that different nucleases may preferentially drive either indel or insertion events.
- The same nucleases from Figure 2 were then screened in HEK 293s for targeted insertion frequency.
- Among the pool of nucleases evaluated, ARCUS01 demonstrated the lowest level of targeted gene insertion (6.07%). On the other hand, ARCUS06 produced 13.9% productive insertions at the cut site (**Figure 4**).

Figure 5. Altering the nuclease, delivery vehicle, and repair template yields significant improvements in PHH targeted gene insertion

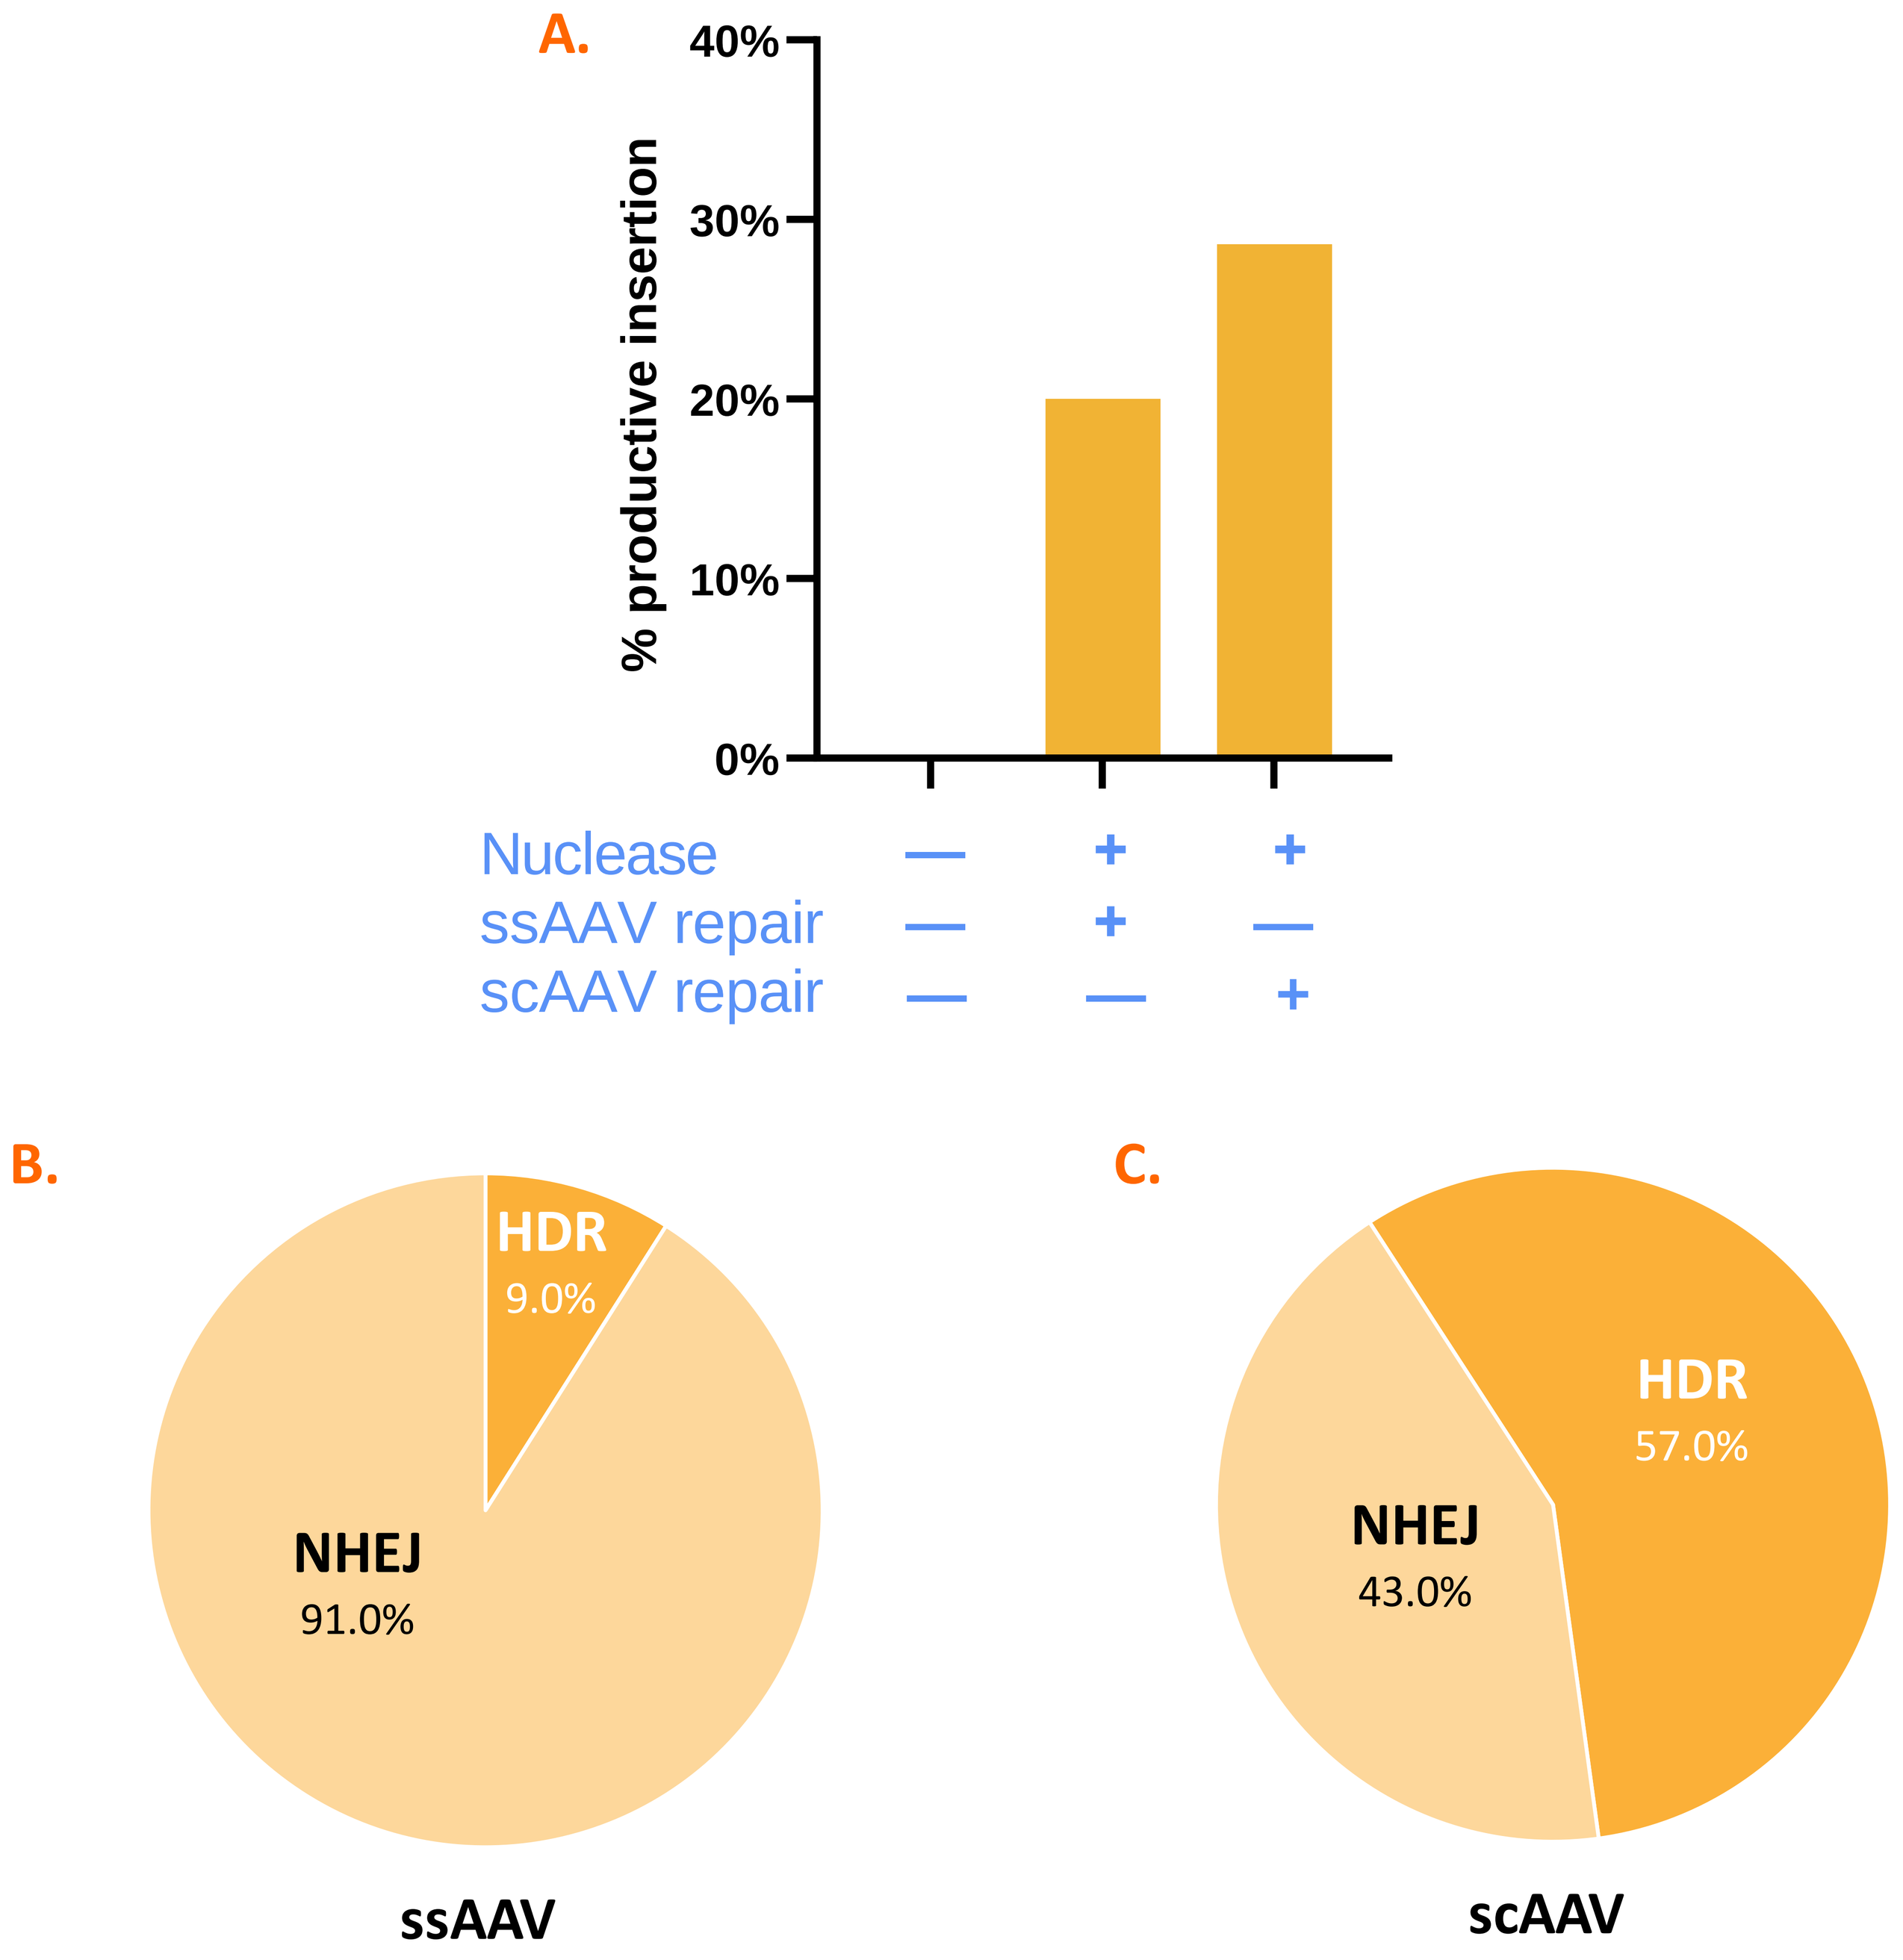
A.
% productive insertion
40%
30%
20%
10%
0%
Nuclease — + +
ssAAV repair — + —
scAAV repair — — +
B.
HDR
9.0%
NHEJ
91.0%
ssAAV
C.
HDR
57.0%
NHEJ
43.0%
scAAV

- Based on the varying degree of insertion frequency observed with different nucleases in HEK 293s, we continued to optimize the nuclease for improved activity. A later-generation nuclease that demonstrated high insertion activity in HEK 293s was subsequently evaluated in PHHs.
- In addition to the higher activity nuclease, we also explored different delivery modalities for both the repair template and the nuclease. We compared gene insertion using both single-stranded AAV (ssAAV) and self-complementary AAV (scAAV) delivery of the repair template. We also utilized LNP as the nuclease delivery tool.
- Using LNP delivery of the optimized nuclease in combination with a scAAV repair template, we observed a total of 28.6% productive insertions, as compared to 20.0% with a ssAAV repair template (**Figure 5A**). Of particular note, 57.0% of the productive insertions were attributed to HDR when scAAV was used (**Figure 5C**), compared to 9.0% when ssAAV was used (**Figure 5B**).

CONCLUSIONS

1. We observed a nuclease-mediated effect on targeted gene insertion, indicating that the ability to screen nucleases for their insertion frequency is imperative when selecting enzymes.
2. Additionally, a scAAV repair template demonstrated an increased ability to insert onto the chromosome via HDR, thus enhancing productive insertion of the transgene.
3. We achieved ~30% productive insertions in non-dividing PHHs (>50% HDR-mediated) using ARCUS, a frequency which to our knowledge has not previously been demonstrated.
4. These data highlight the unique capabilities of the ARCUS gene editing platform and its potential use as an *in vivo* gene editing tool for targeted gene insertion.

USING ARCUS FOR GENE INSERTION

FIGURE 1. Mechanism of targeted gene insertion using ARCUS

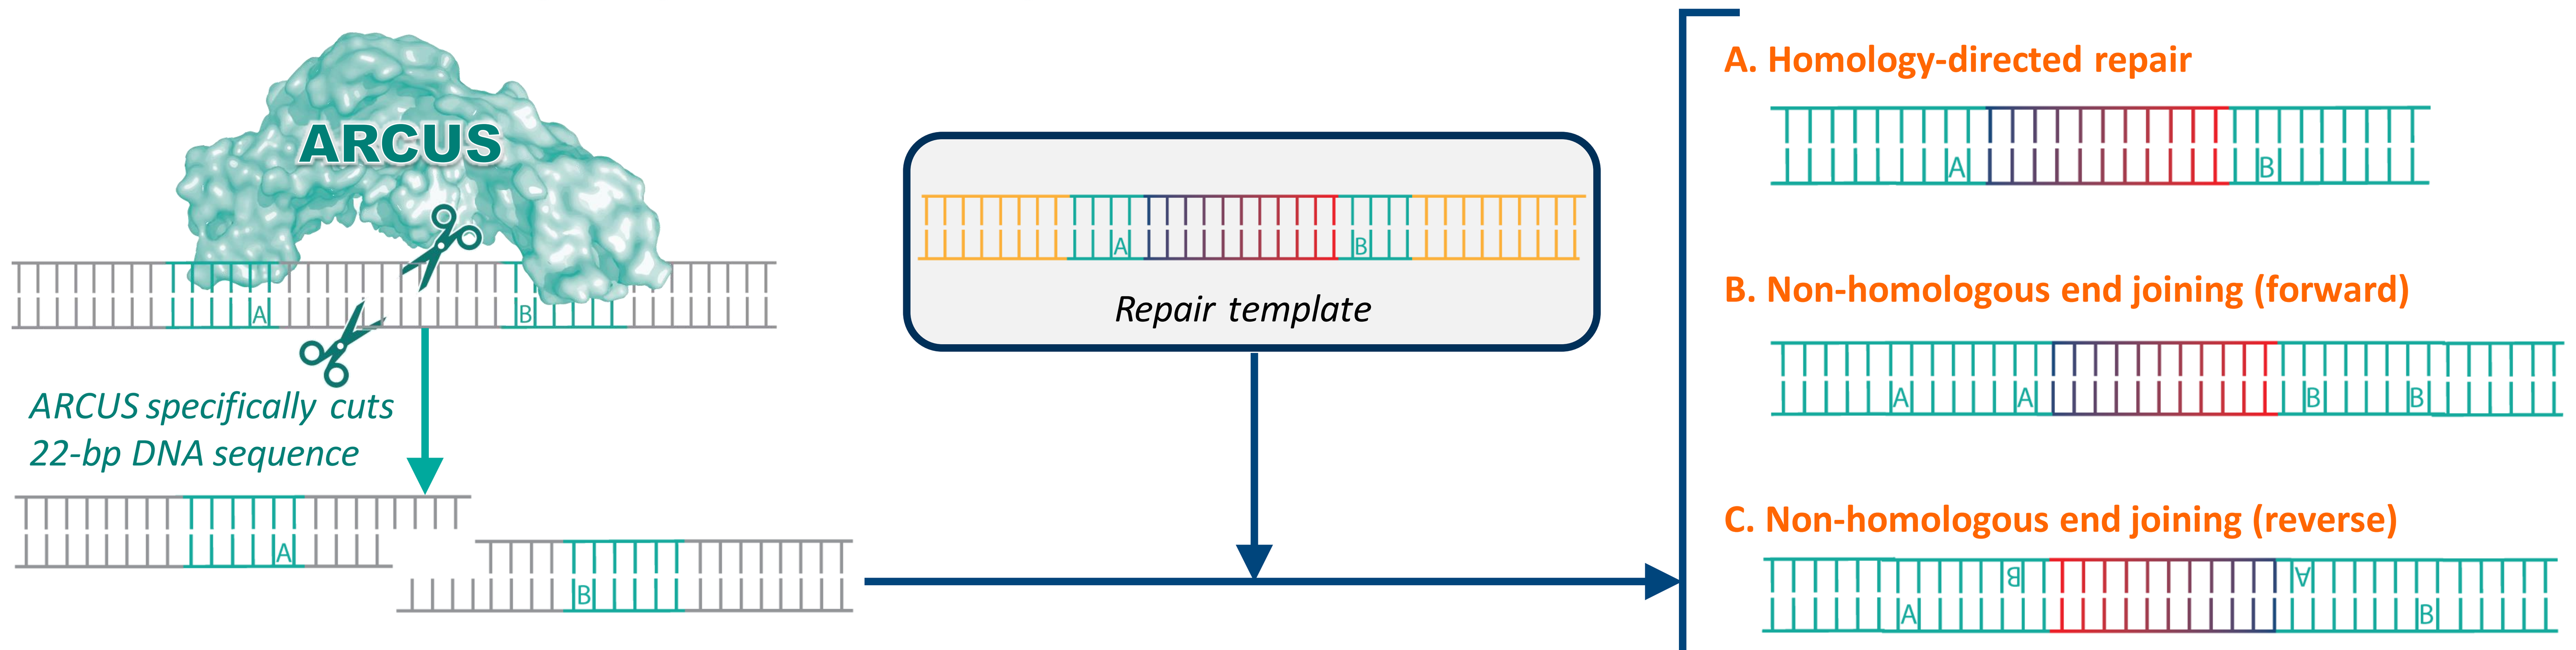
ARCUS
A
B
ARCUS specifically cuts
22-bp DNA sequence
A
B
A
B
Repair template
A. Homology-directed repair
A
B
B. Non-homologous end joining (forward)
A
A
B
B
C. Non-homologous end joining (reverse)
B
A
A
B

PRECISION BIOSCIENCES

Precision Genome Engineering • March 22, 2023 • Whistler, British Columbia, Canada